Bench-to-Bedside Translation of Ciclopirox Prodrug for the Treatment of Non-Muscle Invasive and Muscle-Invasive Bladder Cancer

THE UNIVERSITY OF KANSAS CANCER CENTER

Scott J. Weir[1], Partha Ranjarajan[1], Robyn Wood[1], Karl Schorno[2], Prabhu Ramamoorthy[1], Lian Rajewski[2], Kathy Heppert[2], Michael J. McKenna[3], William McCulloch[4], Greg A. Reed[1], Amanda Brinker[1], Michael J. Baltezor[2], Roy A. Jensen[1], John A. Taylor III[1], Shrikant Anant

NCI Designated Cancer Center

BEST HOSPITALS U.S.News & WORLD REPORT NATIONAL CANCER 2017-18

Abstract #
6774

Abstract

Ciclopirox (CPX) is contained in a number of FDA-approved topical antifungal drug products as the free acid and olamine salt. CPX possesses anticancer activity in a number of *in vitro* and *in vivo* preclinical models. Its clinical utility is limited as an oral anticancer agent, however. The oral bioavailability of CPX is quite low due to extensive first-pass effect. The poor water solubility of CPX and its olamine salt prevent formulation as an injectable drug product. Thirdly, dose-limiting gastrointestinal toxicities were observed following four-times daily oral dosing of CPX in patients with advanced hematologic malignancies. Ciclopirox Prodrug (CPX-POM), in contrast, has demonstrated excellent bioavailability via injectable routes of administration. Here, we describe the preclinical characterization of CPX-POM, a novel anticancer agent being developed for the treatment of non-muscle invasive (NMIBC) and muscle-invasive (MIBC) bladder cancer. Following IV, SQ and IP administration to mice, CPX-POM is rapidly and completely metabolized to CPX in blood via circulating phosphatases. CPX and its major, inactive glucuronide metabolite are extensively eliminated in urine. At well-tolerated doses, steady-state urine concentrations of CPX exceed *in vitro* IC50 values in mice by 15-30 fold. CPX inhibited cell proliferation, colony formation, and bladdosphere formation *in vitro* in T24 (NMIBC) and 253JBV (MIBC) human cell lines in both concentration- and time-dependent manners with IC50 values of 2-4 µM. CPX exposure increased the percentage of NMIBC and MIBC cells arrested at the S and G0/G1 phases, and induced cell death. CPX exposure significantly reduced expression of genes at the mRNA level involved in cancer stem cell signaling pathways including Notch, Wnt, and Hedgehog. CPX was shown to inhibit bladder cancer cell growth *in vitro* by inhibiting the Notch 1 signaling pathway. The validated N-butyl-N-(4-hydroxybutyl) nitrosamine (BBN) chemical carcinogen mouse model of bladder cancer was employed to establish in vivo preclinical proof of principle for CPX-POM. Over the once-daily IP dose range of 25-200 mg/kg, CPX-POM treatment resulted in significant decreases in bladder weight, a clear migration to lower stage tumors, dose-dependent reductions in Ki67 and PCNA staining, as well as a reduction in PCNA-expressing cells. All CPX-POM doses were well tolerated with no evidence of toxicity to the urinary tract based on blinded pathologic evaluation. There were also dose-dependent decreases in Notch 1, Presenilin 1, and Hey 1 in bladder cancer tissues obtained from CPX-POM-treated animals. Tumor response was similar, *in vivo*, following once-daily and three-times weekly CPX-POM administration. CPX-POM has received FDA clearance to proceed to Phase I, and is currently being evaluated in a first-in-human trial in patients with advanced solid tumors.

Background & Significance

Bladder cancer is a devastating disease that currently ranks as the fourth most common cancer among men and the sixth most common among men and women combined. The American Cancer Society estimates that in 2018 alone, 81,190 new cases will be diagnosed in the U.S. and 17,240 will die of the disease. Bladder cancer is two defined as two diseases, each with different treatment approaches and outcomes.

Eighty percent of newly diagnosed patients have non-muscle invasive bladder cancer (NMIBC) (Heney 1992). Despite endoscopic resection followed by topical administration (via bladder instillation) of either immunotherapy (i.e., the Bacillus Calmette-Guerin vaccine) or chemotherapy (e.g., Mitomycin C, cisplatin), 60-70% of NMIBC will recur (Zlotta 2000; Aldousari 2010). Twenty to thirty percent of NMIBC cases progress to MIBC (Zlotta 2000; Aldousari 2010) for which the gold standard treatment is neoadjuvant cisplatin-based chemotherapy followed by radical cystectomy (Millan-Rodriguez 2000; National Comprehensive Cancer Network 2013). Patients who progress from NMIBC to MIBC have a lower survival rate than for those who present with *de novo* MIBC (Grossman 2003).

Urothelial cell carcinomas most commonly occur in the bladder but can involve the upper urinary tract as well. The incidence of upper urinary tract disease ranges from 5-10% (Soloway 1985) and increases with multiple and high risk tumors (Roupret 2013). Delivery of immunotherapy and chemotherapy via bladder instillation fails to reach cancer located in the upper urinary tract. Novel treatment strategies that prevent recurrence and progression of high grade urothelial cell carcinomas are needed (Shabsigh 2009).

Funding

KU THE UNIVERSITY OF KANSAS The Institute for Advancing Medical Innovation

CicloMed

Research presented was supported by The Institute for Advancing Medical Innovation at the University of Kansas Medical Center and CicloMed LLC, Kansas City, MO through a unique public-private partnership.

Innovation

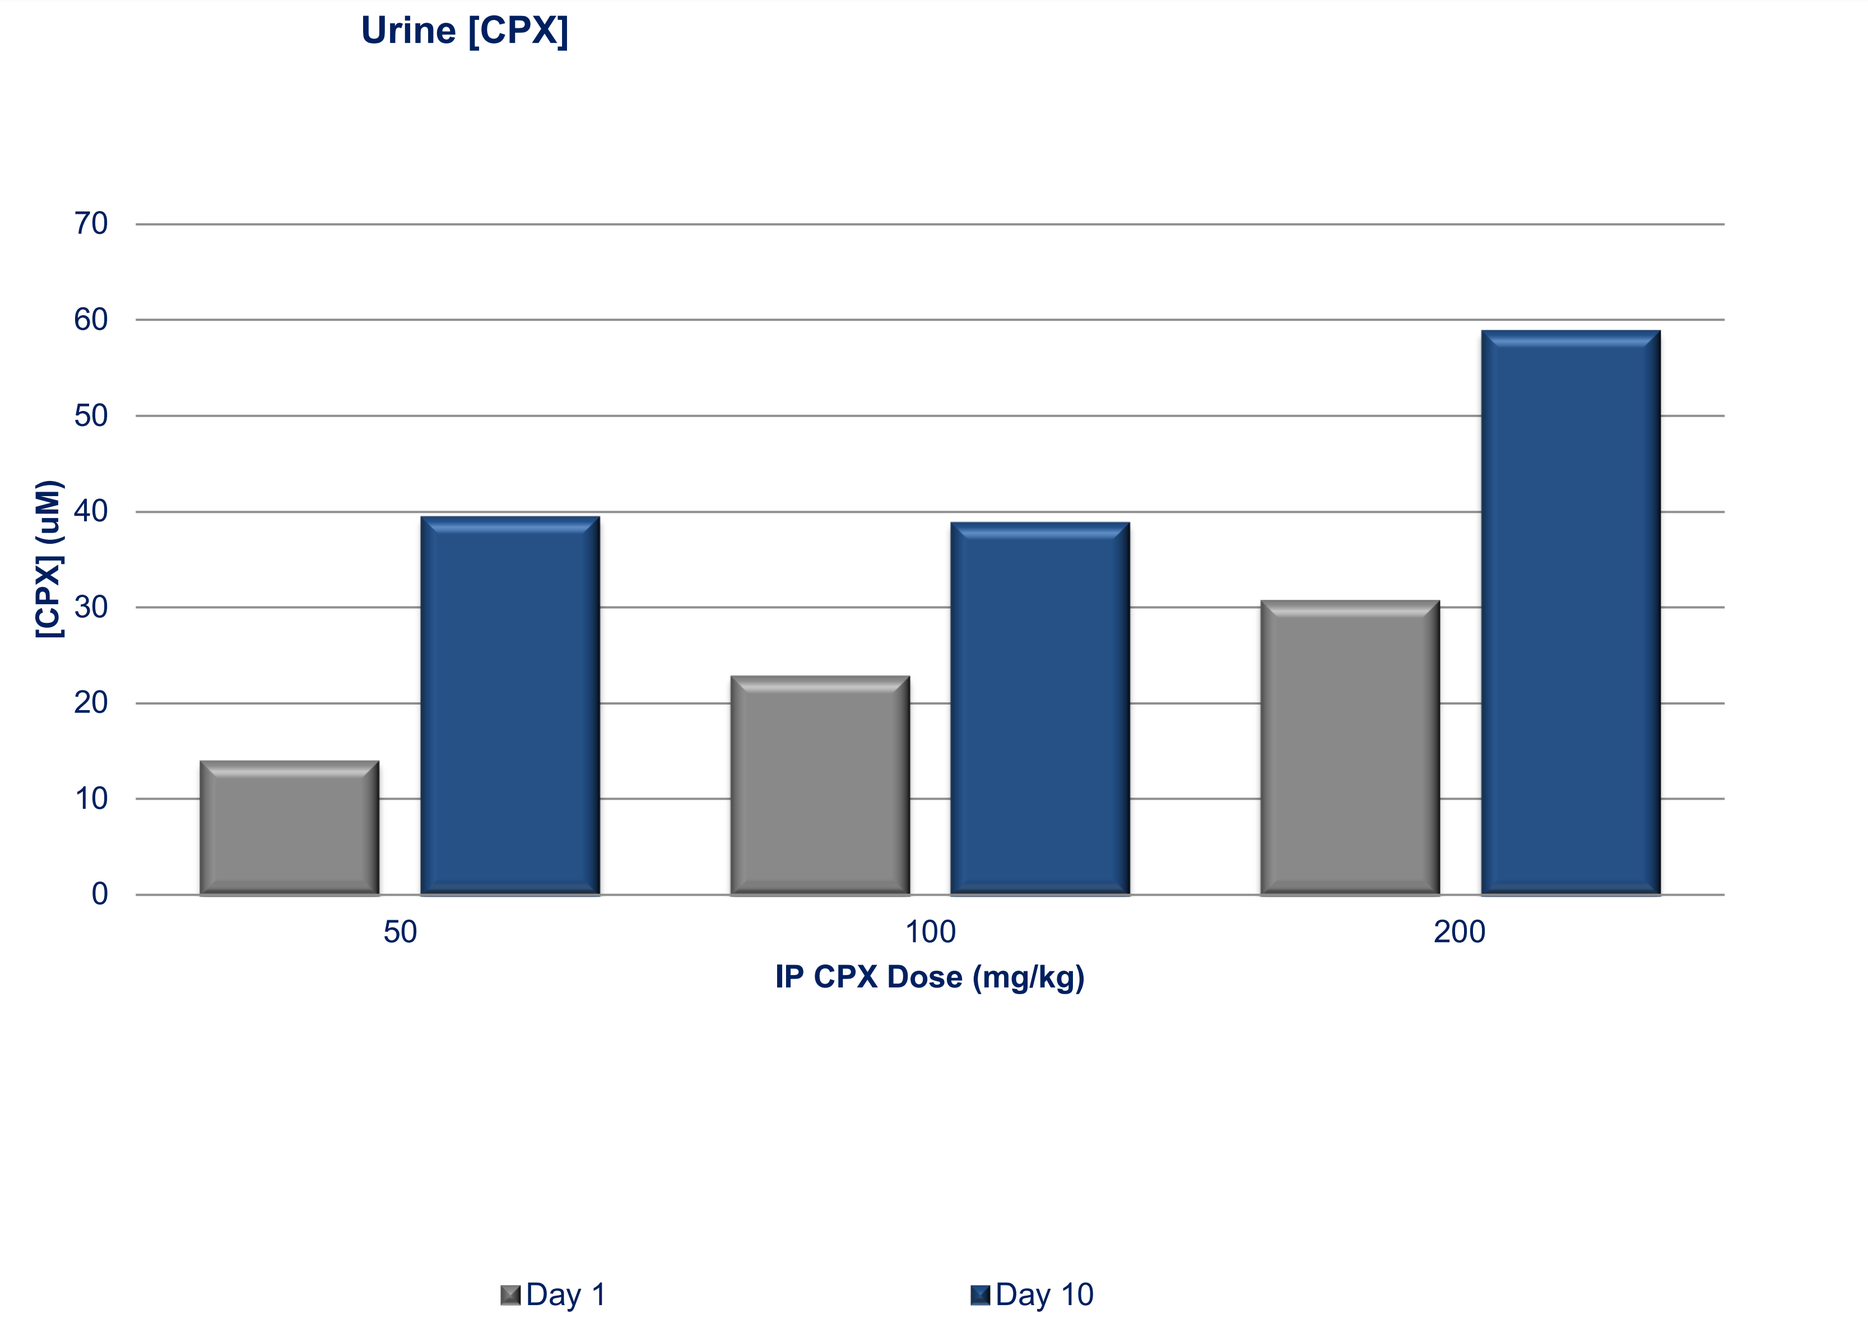
Urine [CPX]
[CPX] (uM)
IP CPX Dose (mg/kg)
Day 1
Day 10

Figure 1. Systemic administration of Ciclopirox Prodrug (CPX-POM) results in selective delivery of the active metabolite, ciclopirox (CPX), to the entire urinary tract. CPX-POM is rapidly and completely metabolized via circulating phosphatases to CPX. CPX is excreted in urine and metabolized to the inactive glucuronide metabolite, that is also eliminated in urine. A portion of the inactive glucuronide metabolite is hydrolyzed in urine of bladder cancer patients to reactivate CPX due to high β-glucuronidase activity.

In Vitro Proof of Principle

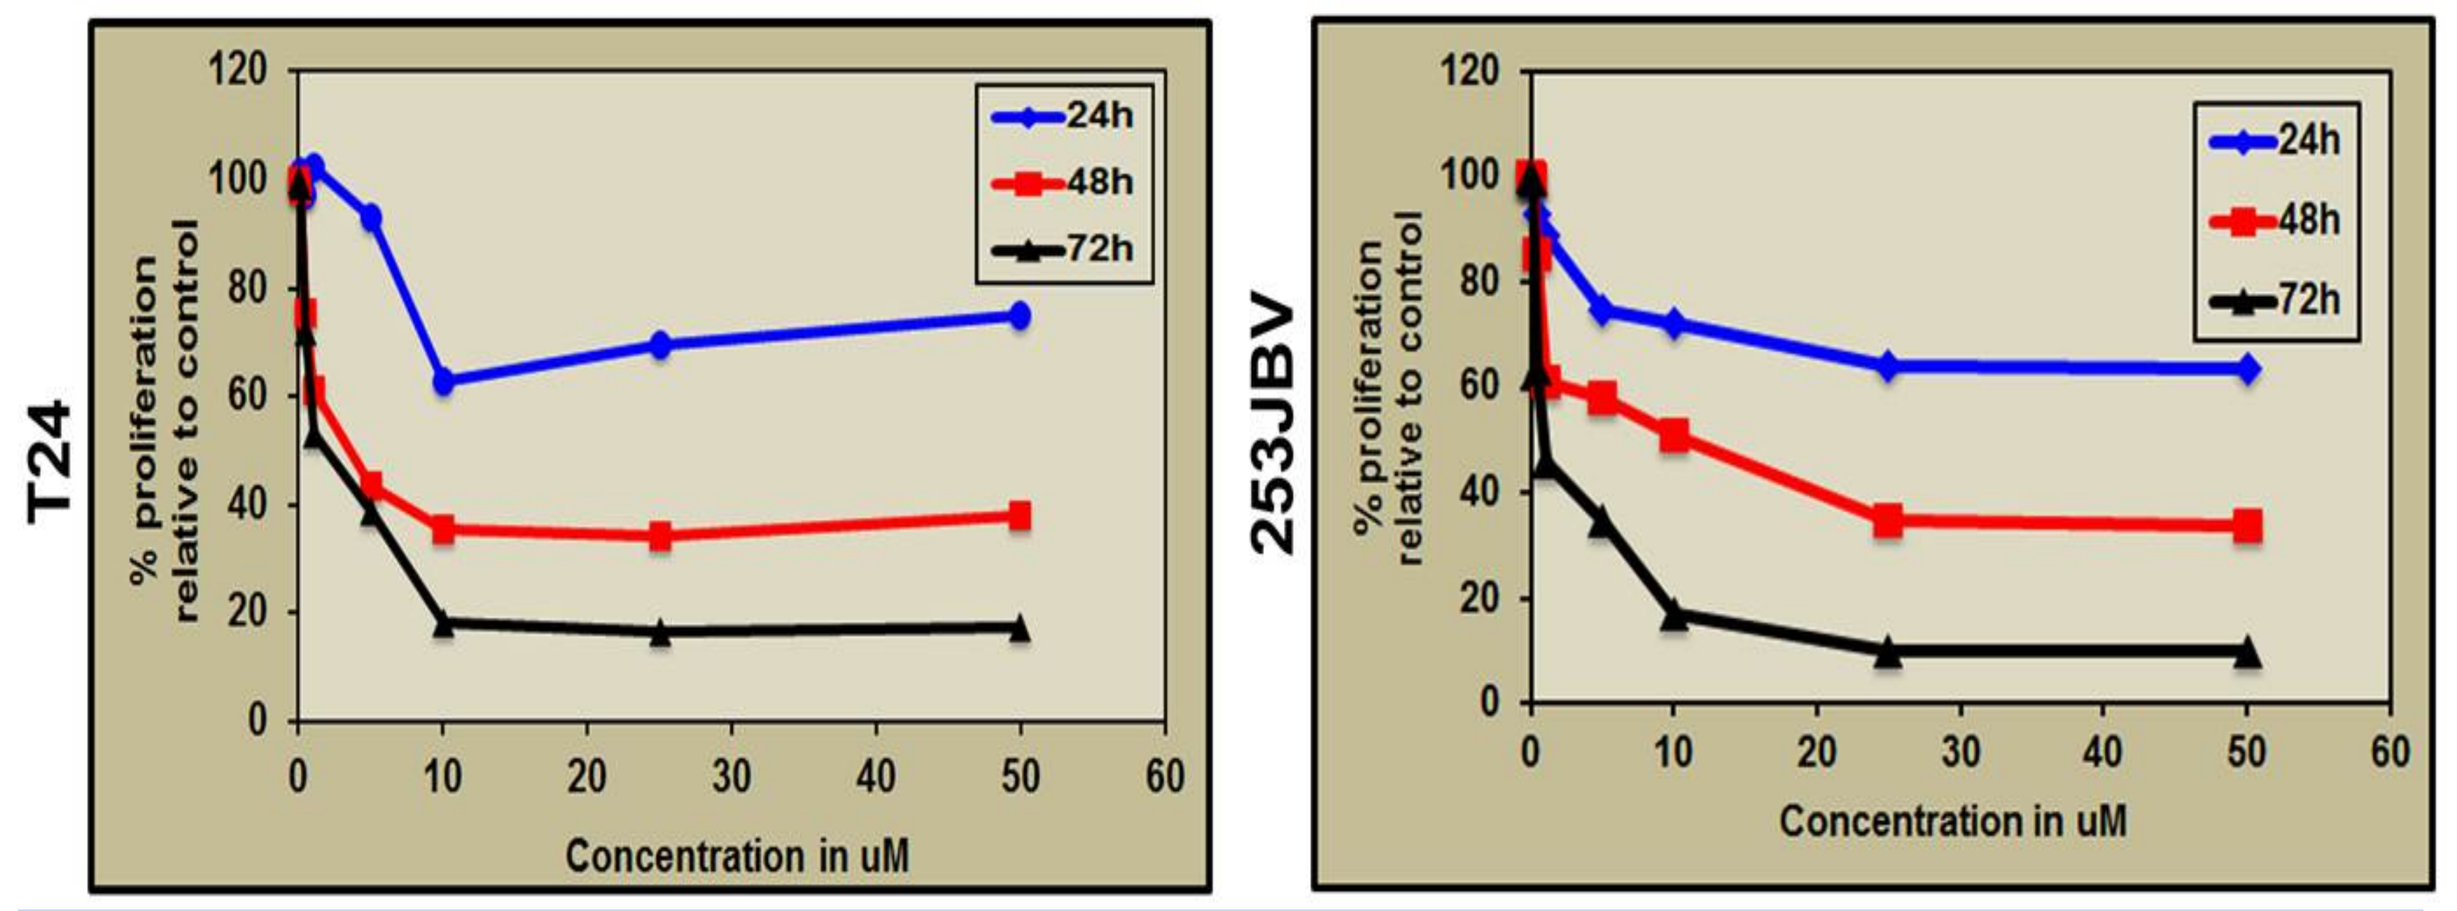
T24
253JBV
% proliferation relative to control
Concentration in uM
24h
48h
72h

Figure 2. CPX inhibited cell proliferation in high grade non-muscle invasive (T24) and muscle invasive (253JBV) bladder cancer cell lines in a concentration-dependent and time-dependent manner with IC50 values of 2-4 µM and 1.0-1.5 µM at 48-hour and 72-hour incubations respectively.

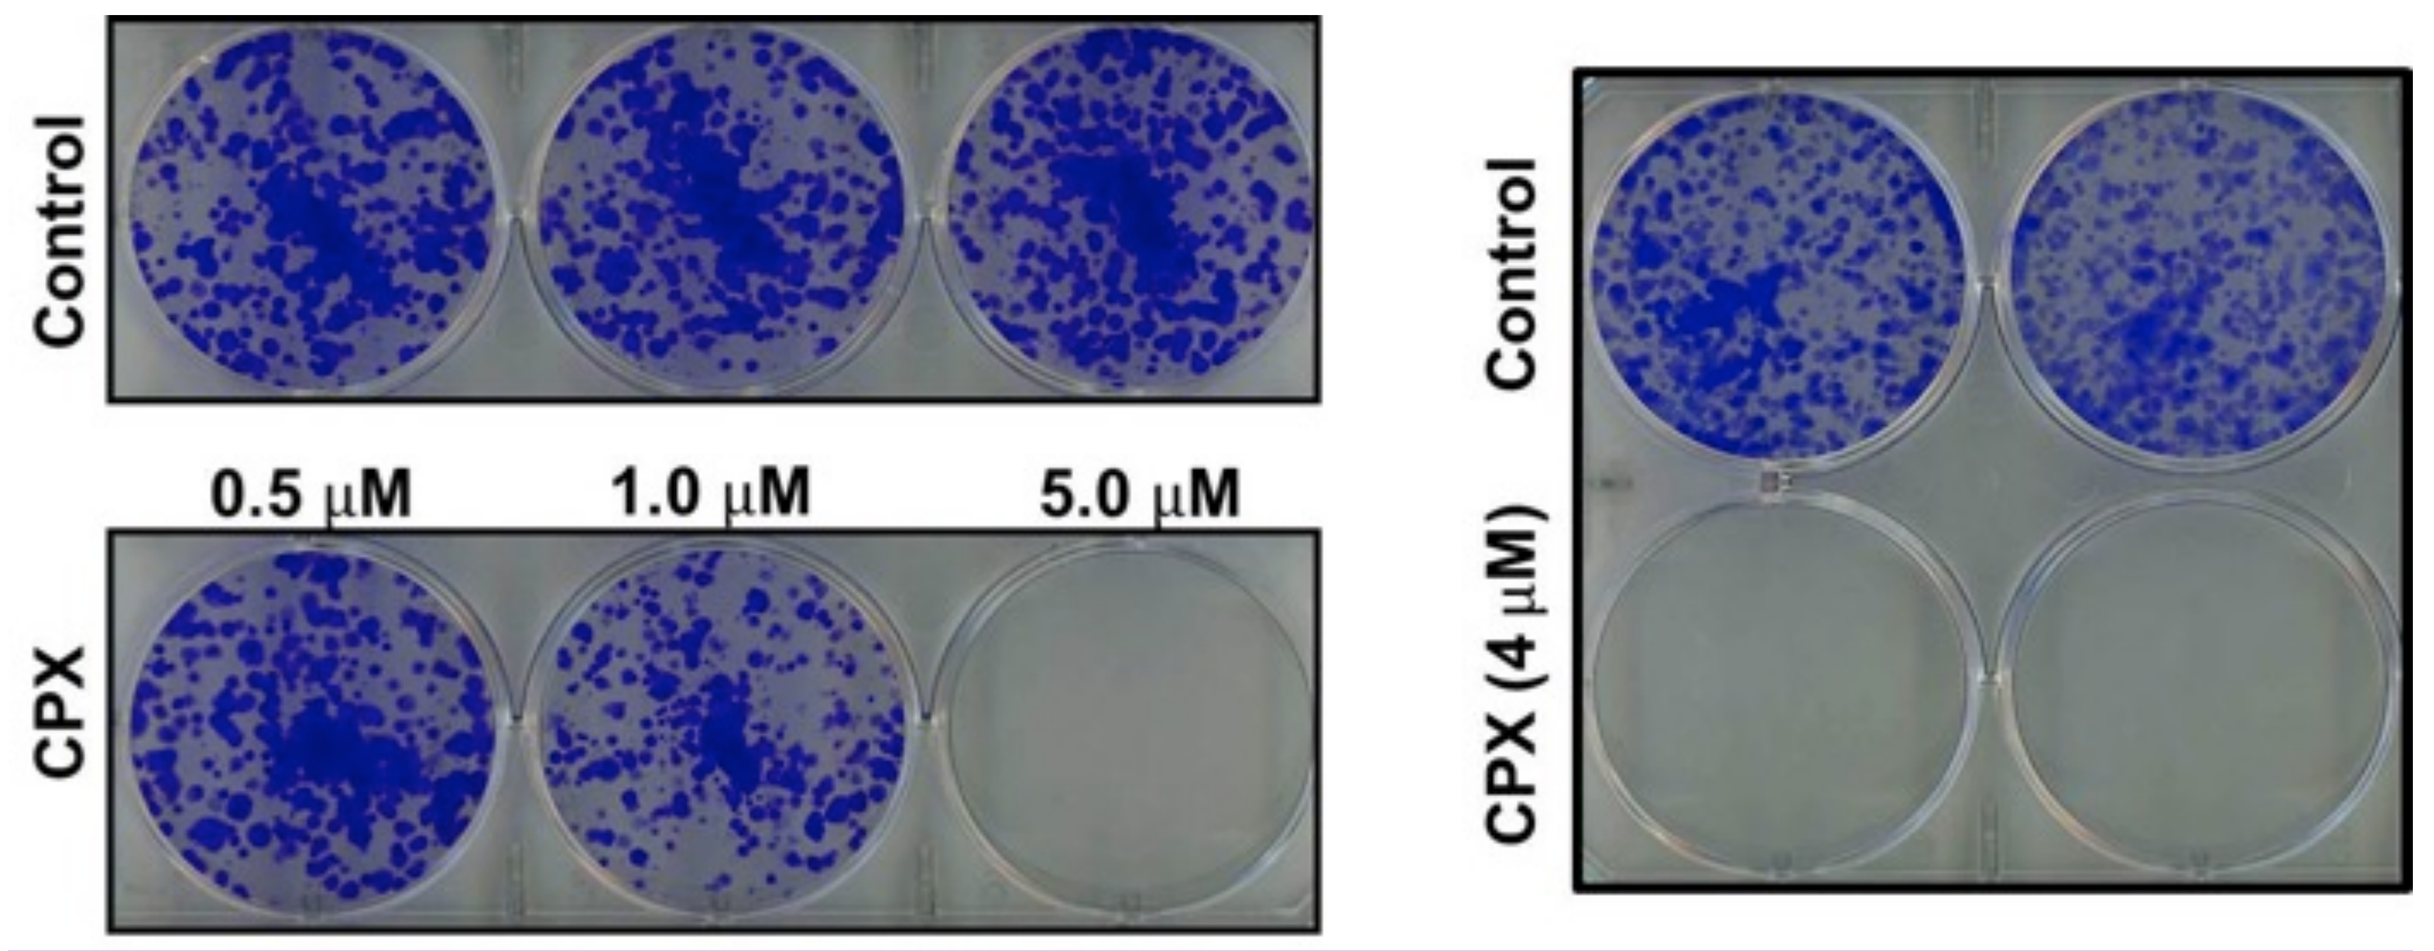
Control
CPX
0.5 µM
1.0 µM
5.0 µM
Control
CPX (4 µM)

Figure 3. CPX exposure reduced the number and size of T24 colonies formed in clonogenicity/colony formation assays.

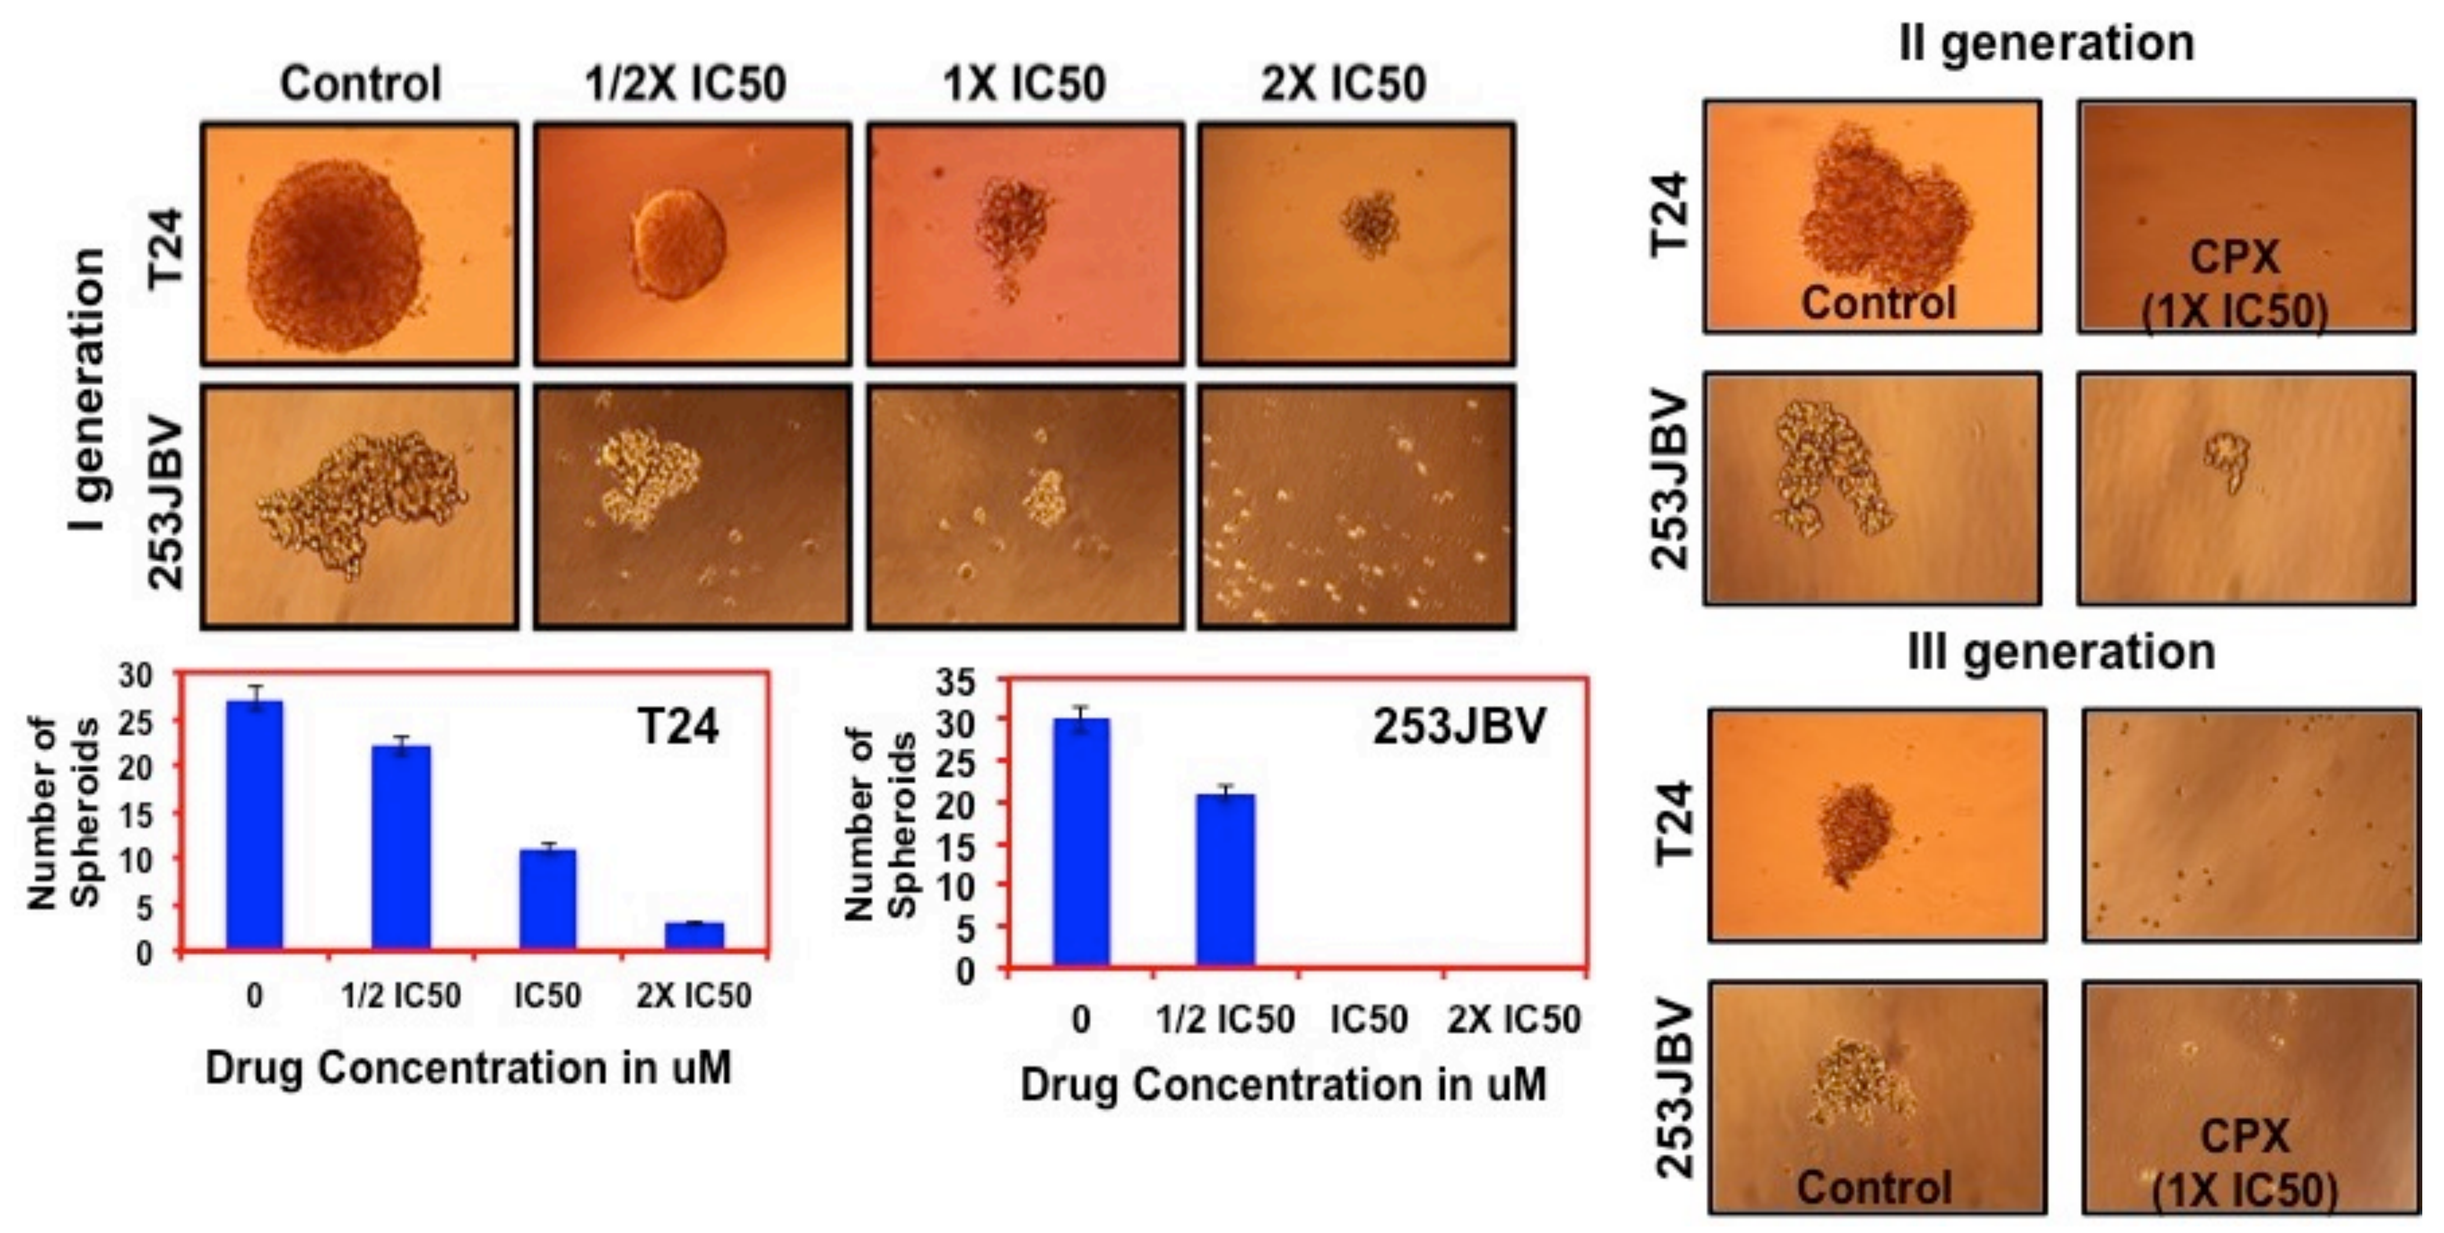
Control
1/2X IC50
1X IC50
2X IC50
I generation
T24
253JBV
II generation
III generation
CPX (1X IC50)
Number of Spheroids
Drug Concentration in uM
0
1/2 IC50
IC50
2X IC50

Figure 4. In the bladdosphere assay, CPX exposure resulted in decreased spheroid size and number of spheroids. T24 and 253JBV second and third generation spheroids were also inhibited as a result of exposure to first generation spheroids. Not shown here, At CPX exposure at 2 µM resulted in an increased percentage of T24 cells arrested at the S and G0/G1 phases of the cell cycle. Also not shown, CPX induced apoptosis, inhibited both invasion and migration via Scratch Plate and Boyden Chamber assays.

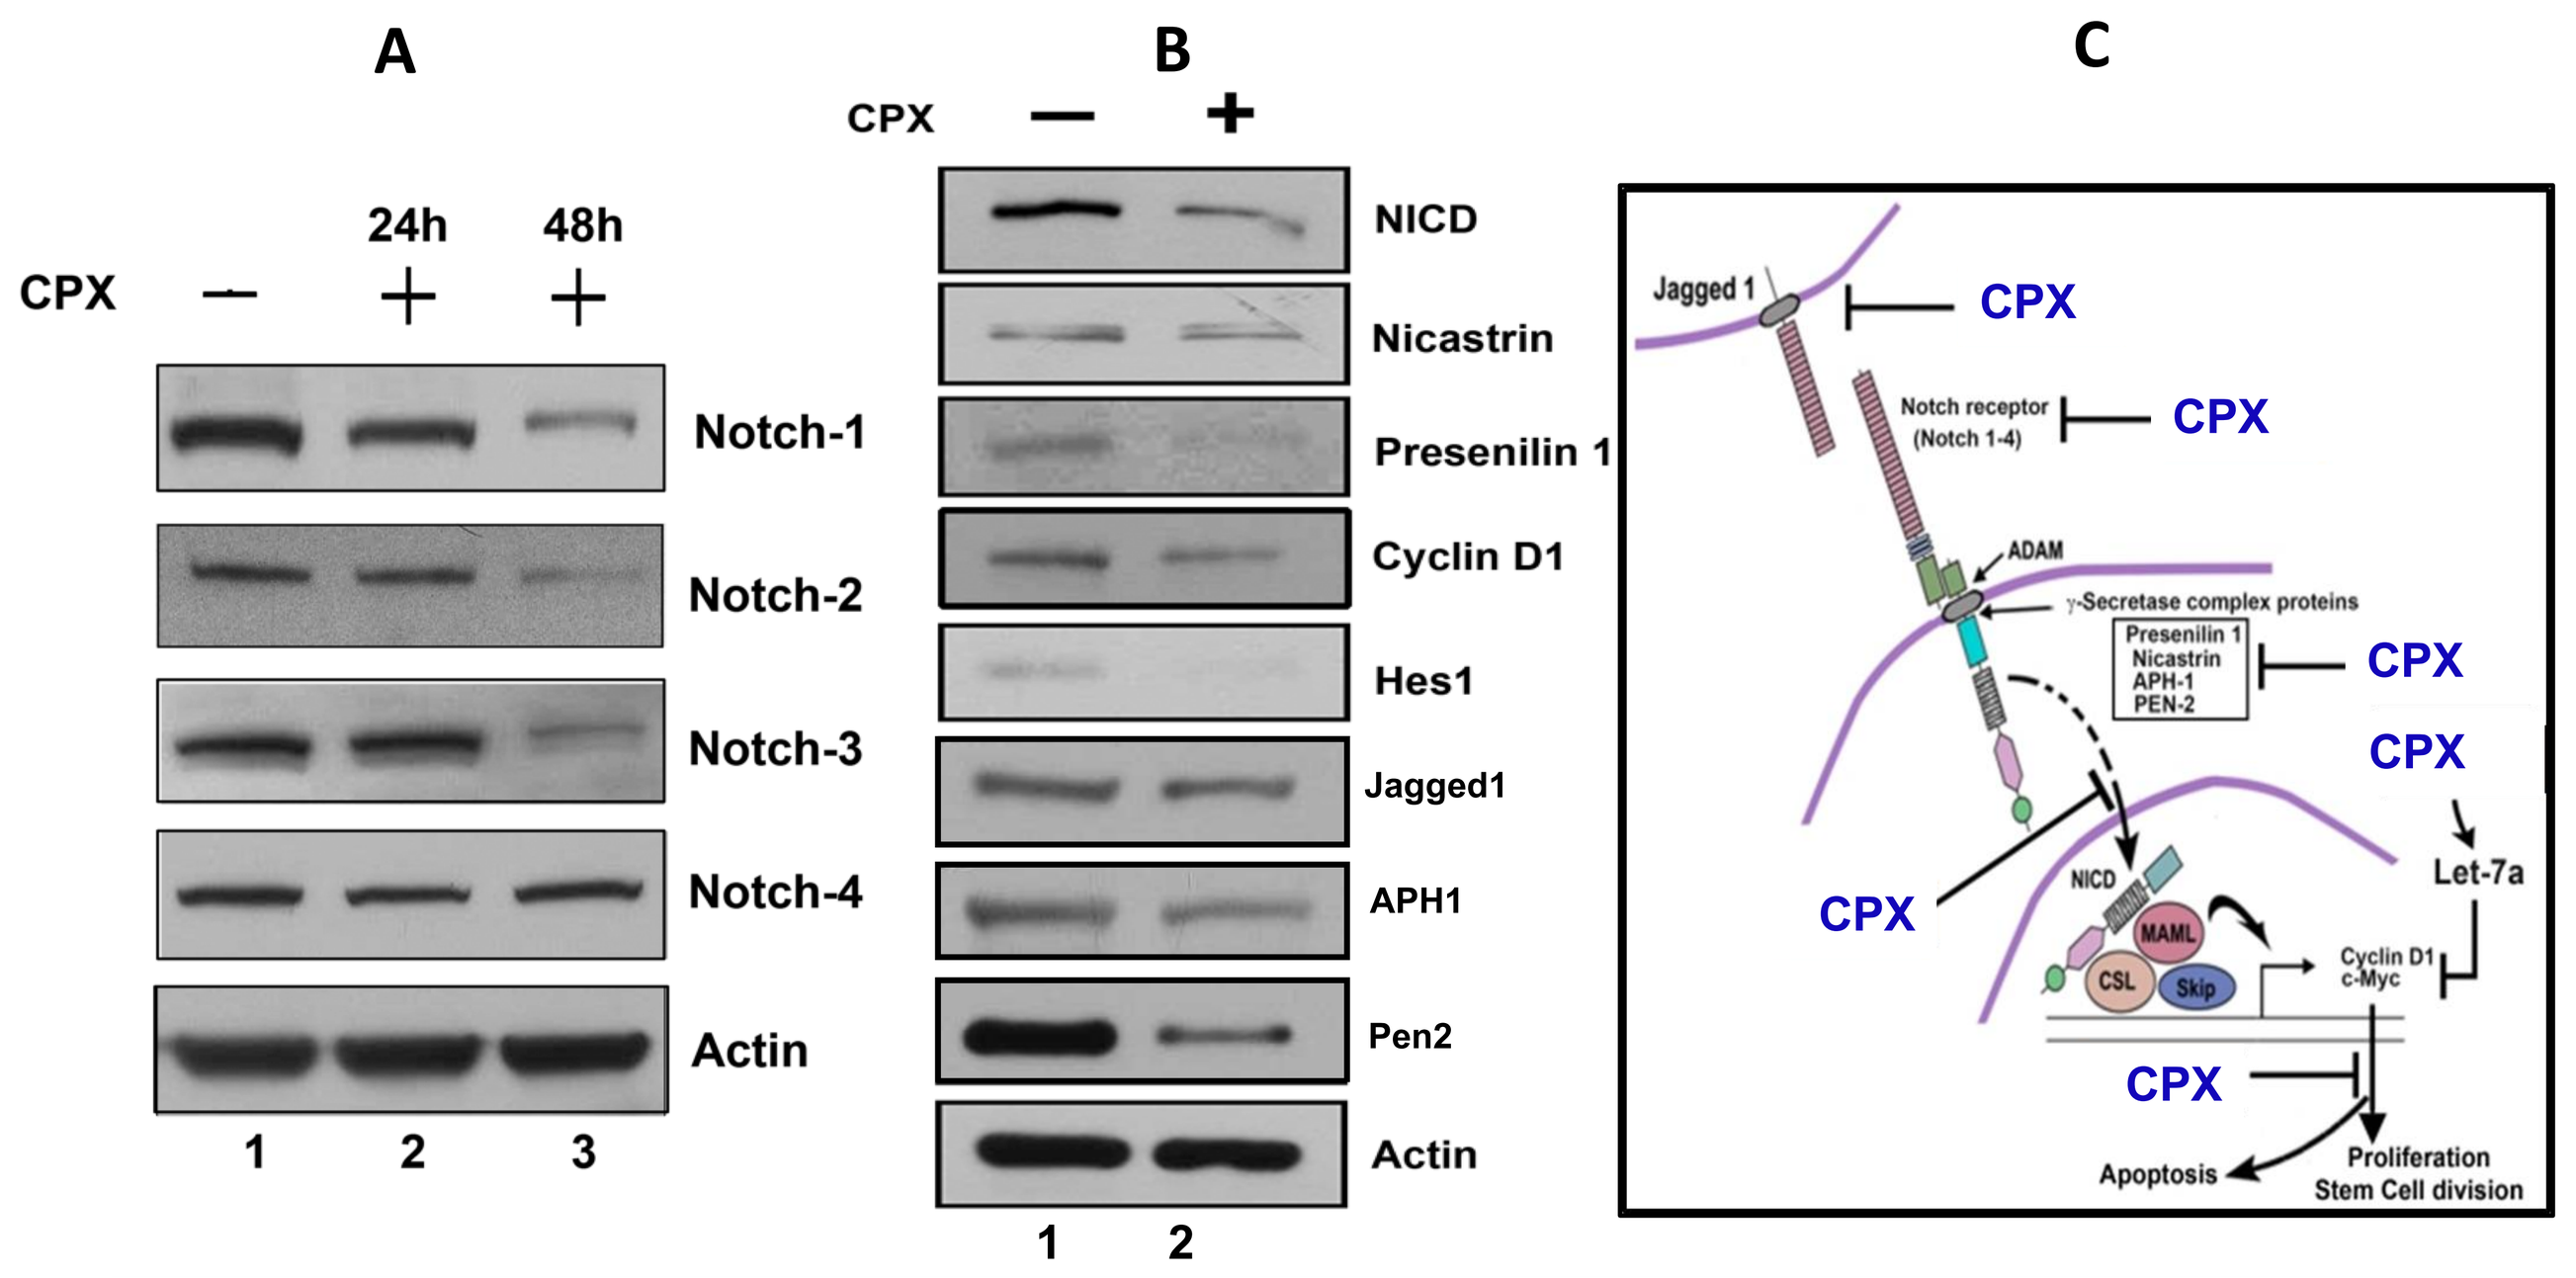
A
B
C
CPX
24h
48h
Notch-1
Notch-2
Notch-3
Notch-4
Actin
NICD
Nicastrin
Presenilin 1
Cyclin D1
Hes1
Jagged1
APH1
Pen2
Jagged 1
Notch receptor (Notch 1-4)
ADAM
γ-Secretase complex proteins
Presenilin 1
Nicastrin
APH-1
PEN-2
Let-7a
NICD
MAML
CSL
Skip
Cyclin D1
c-Myc
Apoptosis
Proliferation
Stem Cell division

Figure 5. CPX inhibited gene expression for Wnt, Hedgehog and Notch cell signaling pathways in T24 and 253JBV cell lines. Protein expression levels for Notch 1, 2 and 3 isoforms were inhibited following exposure to CPX in T24 cells, whereas Notch 4 was not affected (A). CPX inhibited Notch 1 activation and Jagged 1, a ligand for the Notch 1 receptor (B). Additionally, CPX inhibited expression of secretase complex proteins including Presenelin 1, Nicastrin, APH-1, and PEN-2, resulting in decreased levels of cleaved NICD1 reflected in schematic C.

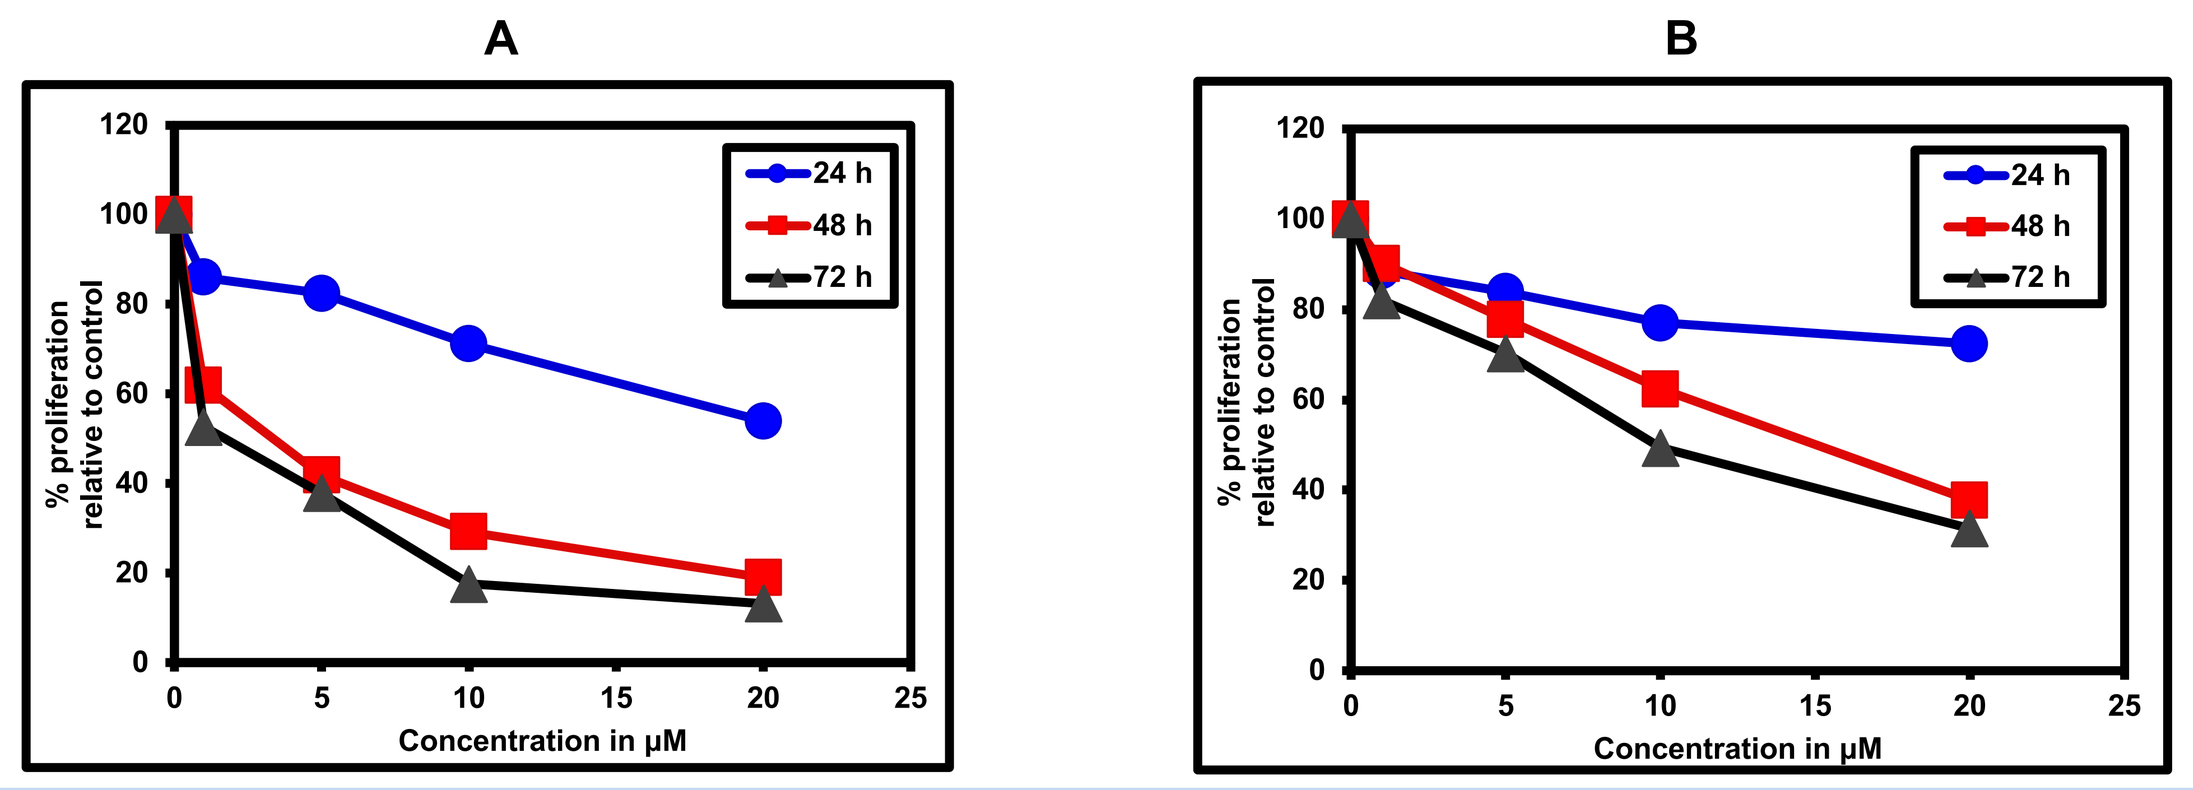
A
B
% proliferation relative to control
Concentration in µM
24 h
48 h
72 h

Figure 6. In T24 cells transfected with a plasmid encoding the Notch Intracellular Domain 1 (NICD1) resulting in overexpression of Notch 1, the antiproliferative effects of CPX were reversed.

In Vivo Proof of Principle

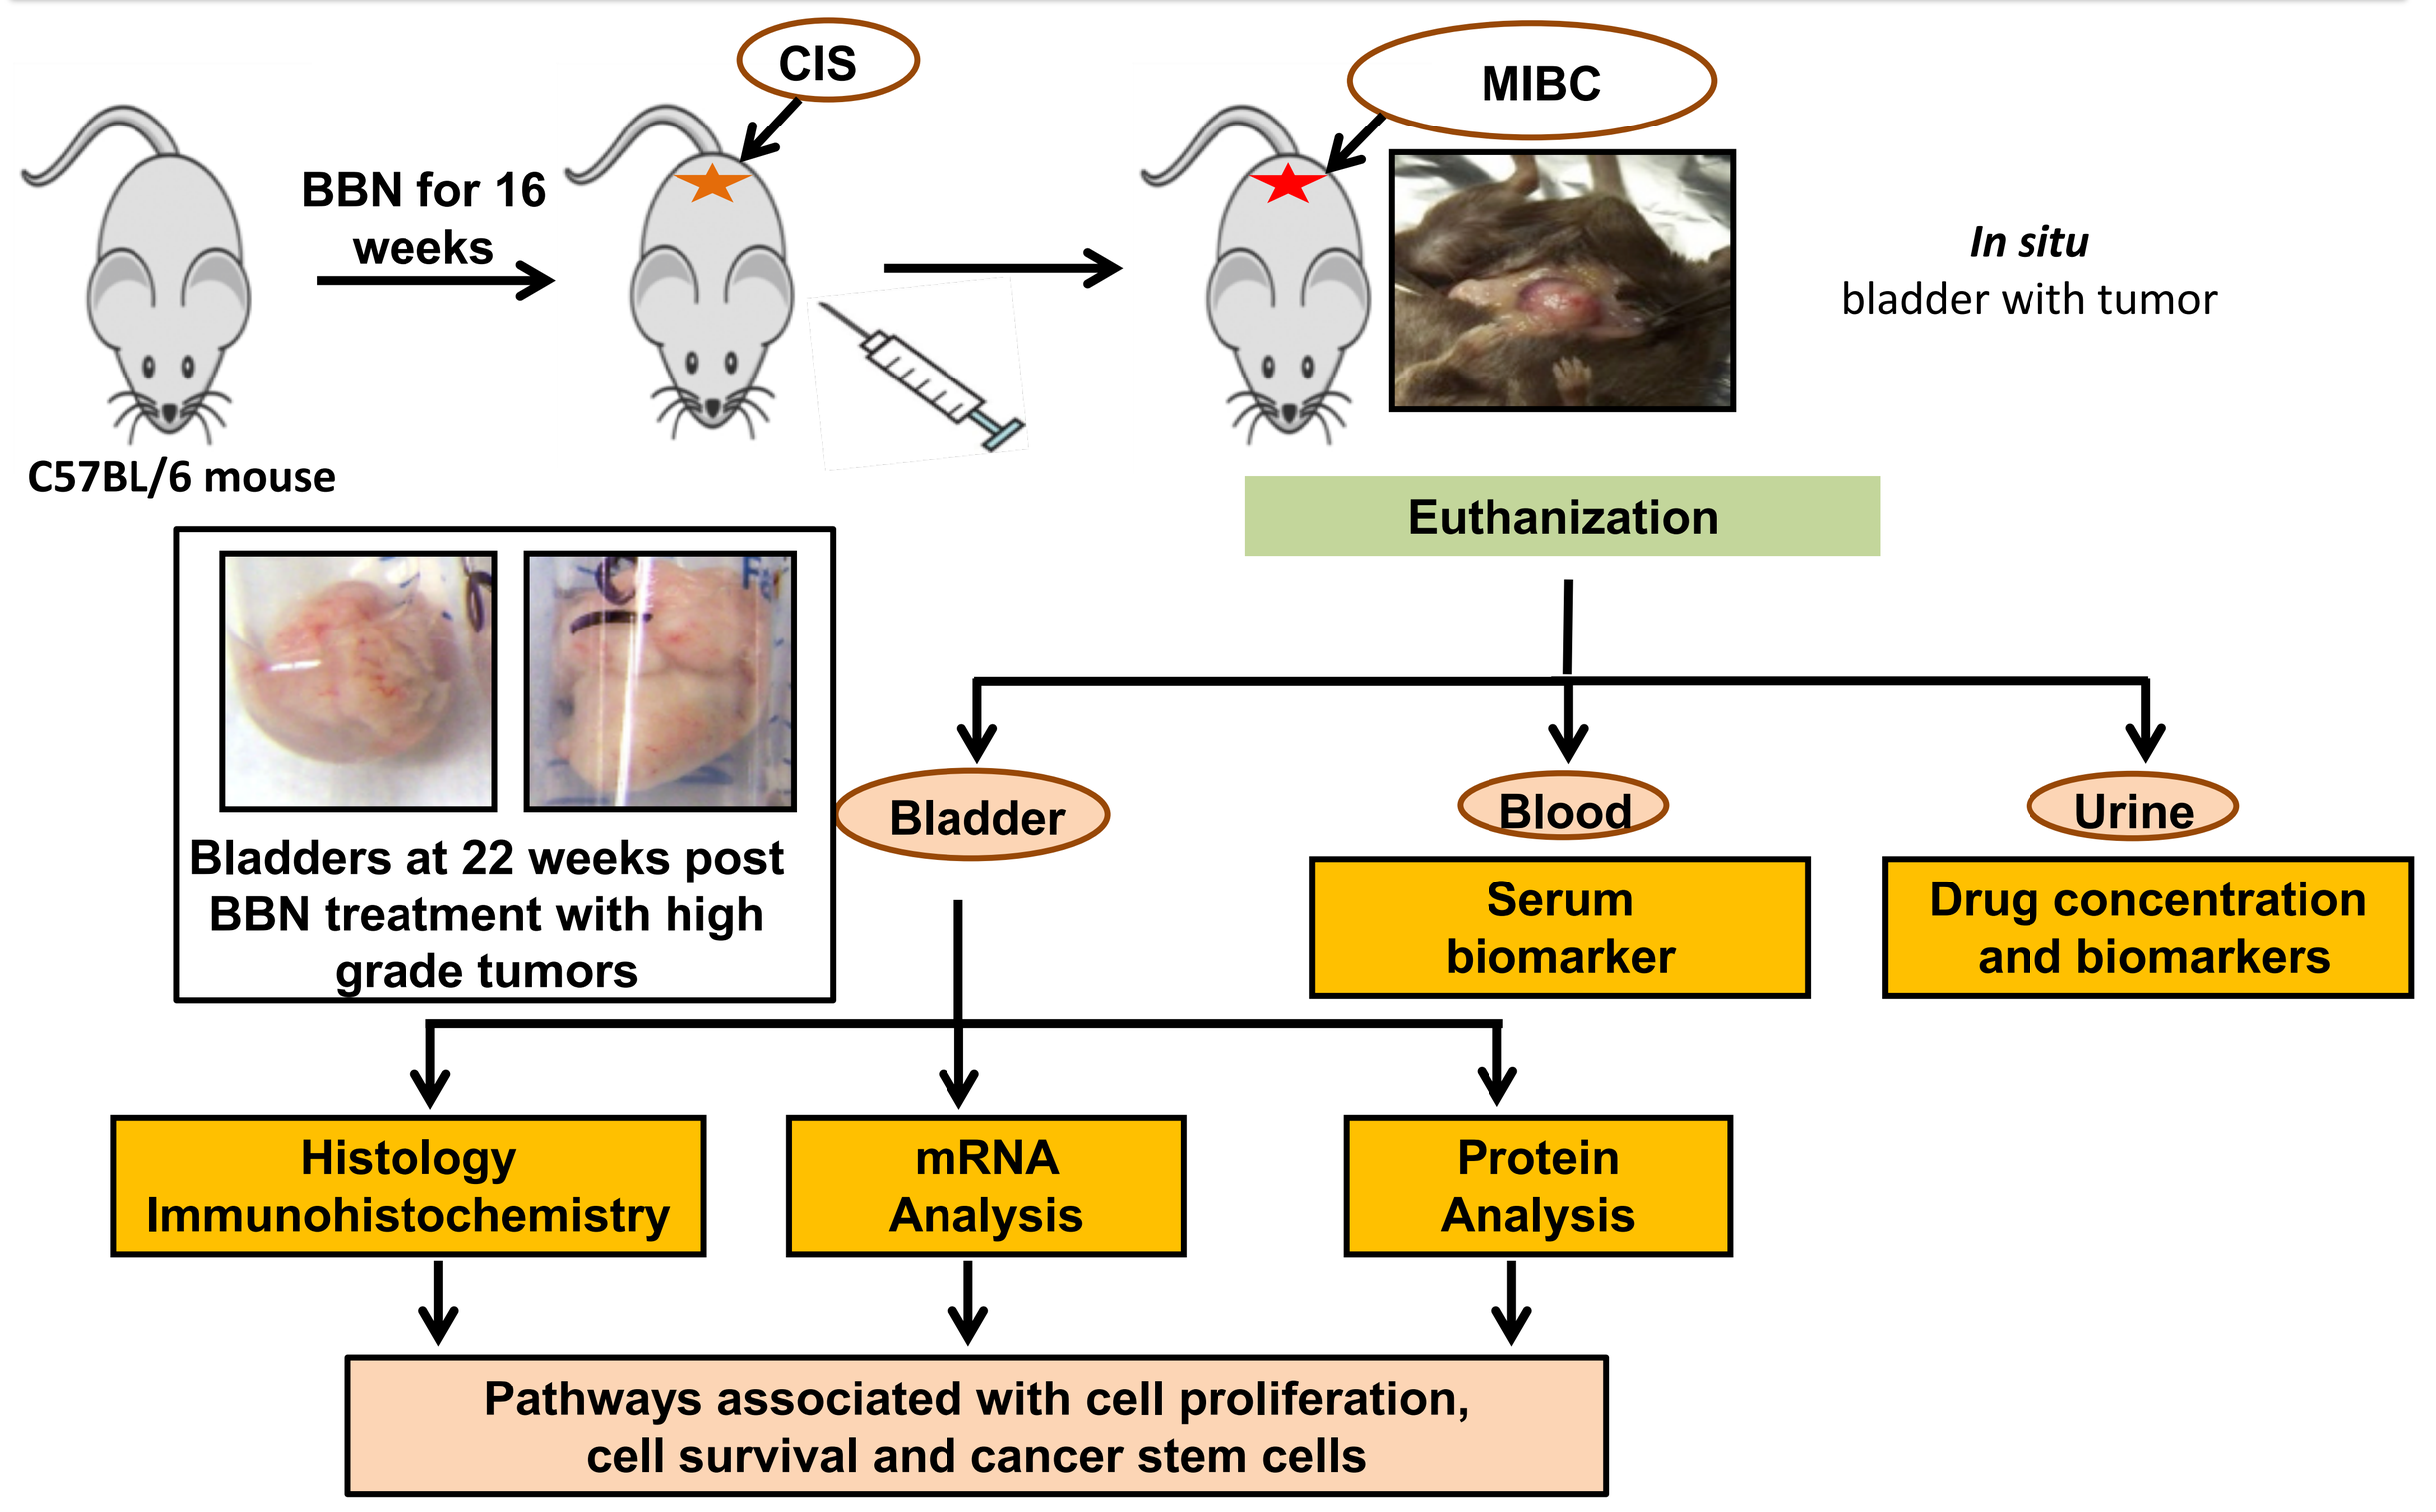
C57BL/6 mouse
BBN for 16 weeks
CIS
MIBC
In situ
bladder with tumor
Euthanization
Bladders at 22 weeks post BBN treatment with high grade tumors
Bladder
Blood
Urine
Serum biomarker
Drug concentration and biomarkers
Histology Immunohistochemistry
mRNA Analysis
Protein Analysis
Pathways associated with cell proliferation, cell survival and cancer stem cells

Figure 7. *In vivo* proof of principle was determined employing the validated N-butyl-N-(4-hydroxybutyl)-nitrosamine (BBN) chemical carcinogen mouse model of bladder cancer. In C57Bl/6 mice, this model demonstrates Carcinoma *in situ* following exposure to BBN for 16 weeks via the drinking water. At 16 weeks, the carcinogen is removed and drug treatment initiated. Without treatment, Carcinoma *in situ* progresses to MIBC over 16-20 weeks. We defined proof of principle for CPX-POM as demonstrating > 30% difference in bladder weight (as a measure of tumor size and volume), >20% difference in tumor stage and grade, and significant changes in cell proliferation as measured by Ki67 and PCNA staining.

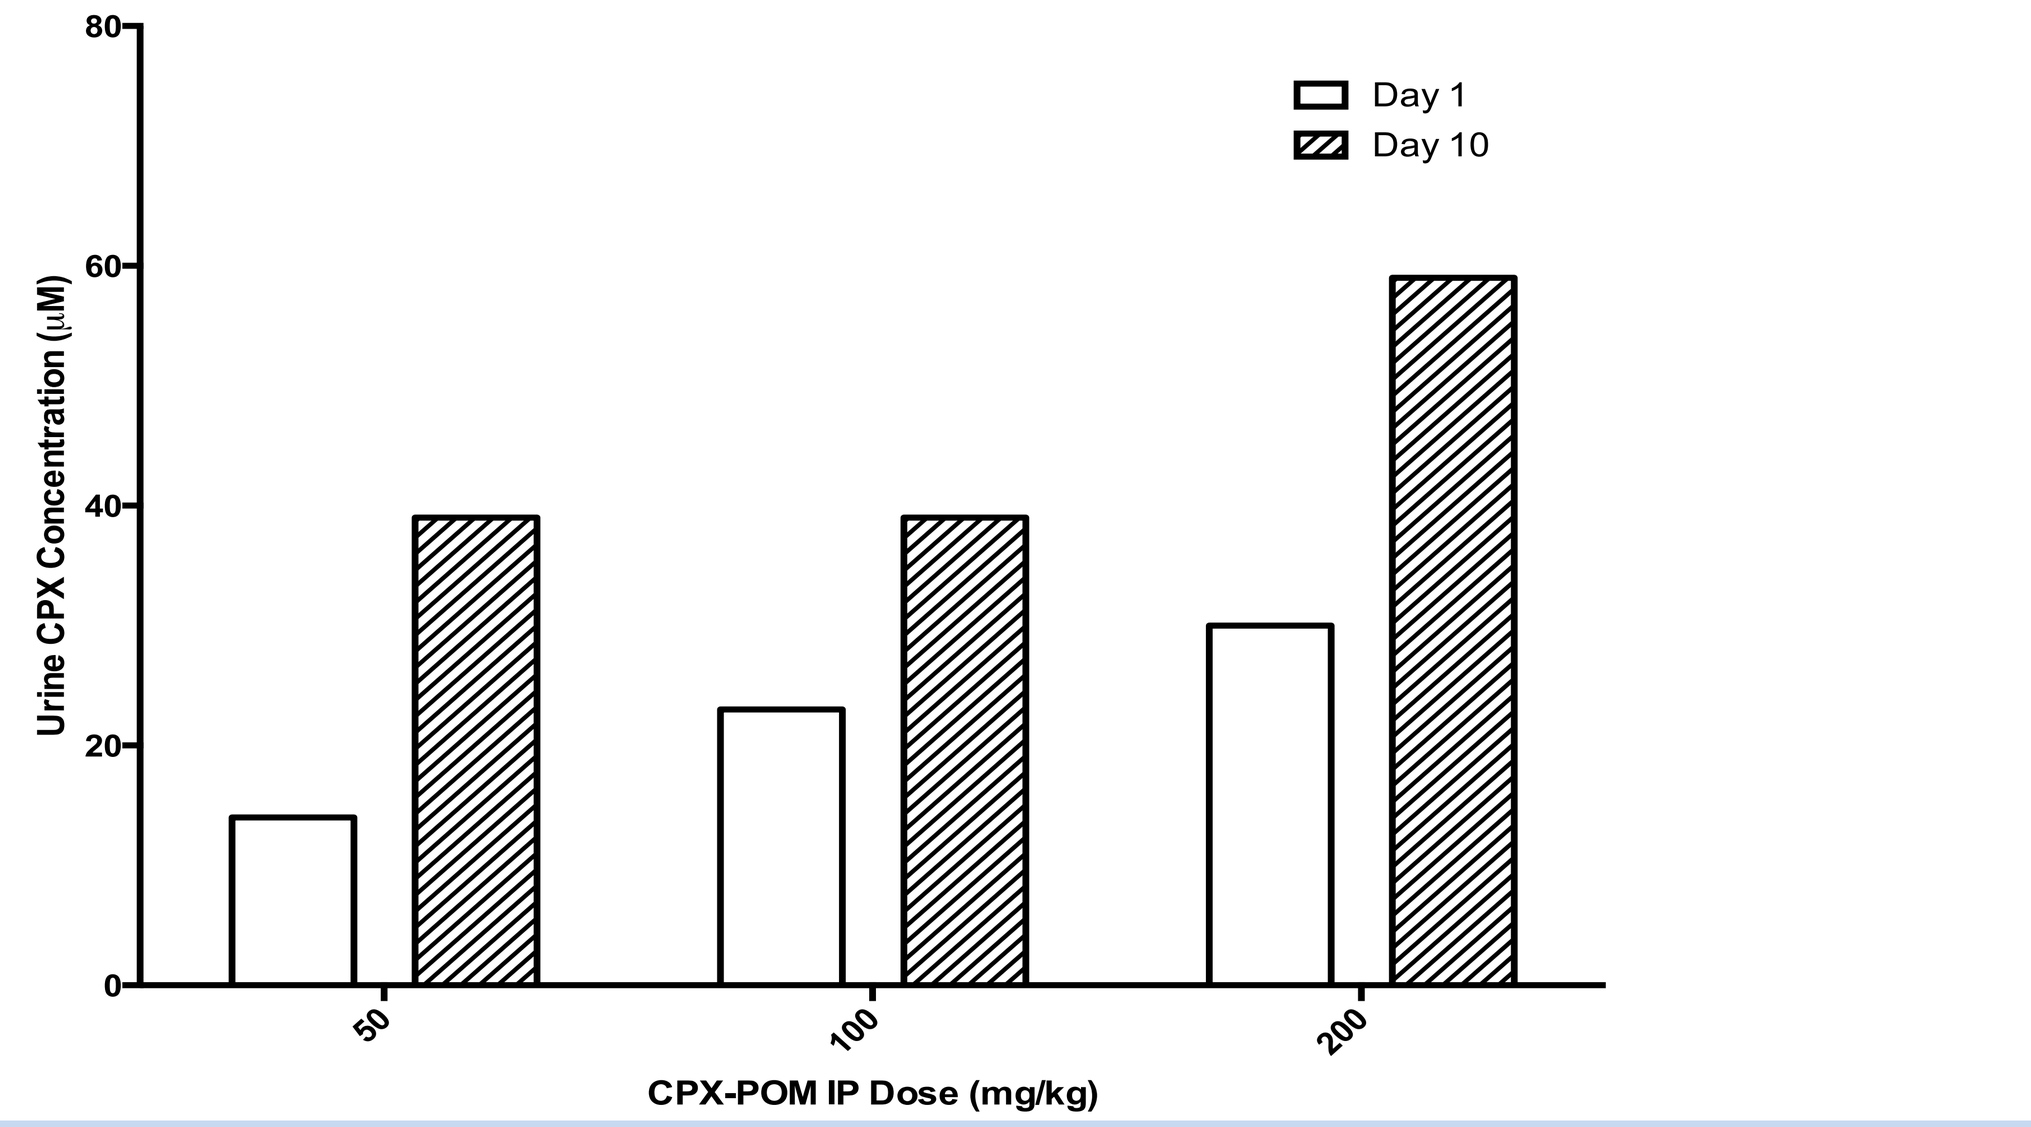
Urine CPX Concentration (µM)
CPX-POM IP Dose (mg/kg)
Day 1
Day 10

Figure 8. Not shown, the absolute bioavailability of CPX following IP CPX-POM was 61%. CPX-POM was not detected in plasma nor urine. The average 24-hour urine CPX concentration over the IP dose range of 50-200 mg/kg exceeded *in vitro* IC50 values (described in Figure 2) by 15 to 60 fold. The once daily IP CPX-POM maximum tolerated dose (MTD) was defined 200 mg/kg.

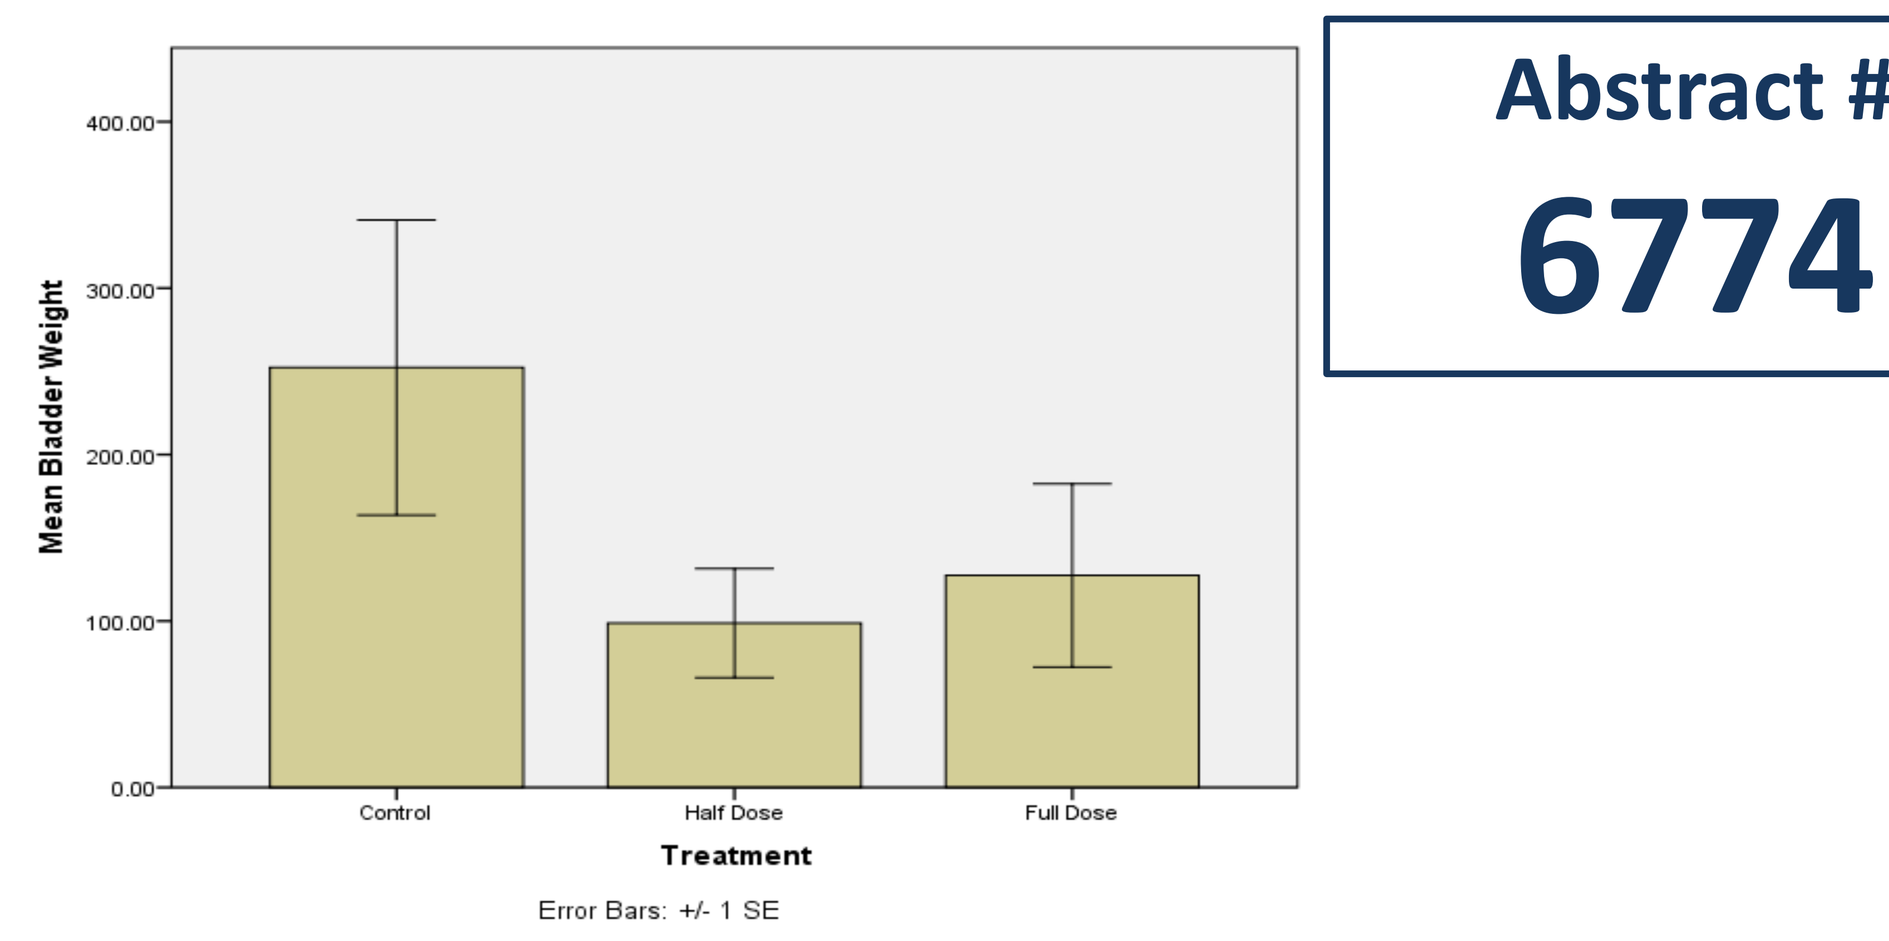
Mean Bladder Weight
Control
Half Dose
Full Dose
Treatment
Error Bars: +/- 1 SE

Figure 9. In the first BBN study, CPX-POM proof of principle was determined at MTD (N=18, 200 mg/kg) and one-half MTD (N=20,100 mg/kg) IP CPX-POM administration resulted in significantly ($p < 0.05$) lower bladder weights compared to control animals. Not shown, there was a moderate to strong correlation between CPX-POM treatment and pathologic outcome (Pearson's Chi-Square R^2=0.74) that did not reach statistical significance (p=0.12). Cell proliferation by Ki67 and PCNA staining was reduced in CPX-POM treated animals. Dose-dependent reductions in Notch 1, Presenelin 1, and Hey 1 in bladder cancer tissues were also observed in CPX-POM treated animals. There were no differences in bladder weight, tumor stage, nor tissue markers of cell proliferation or Notch pathway signaling between the two CPX-POM treatment groups.

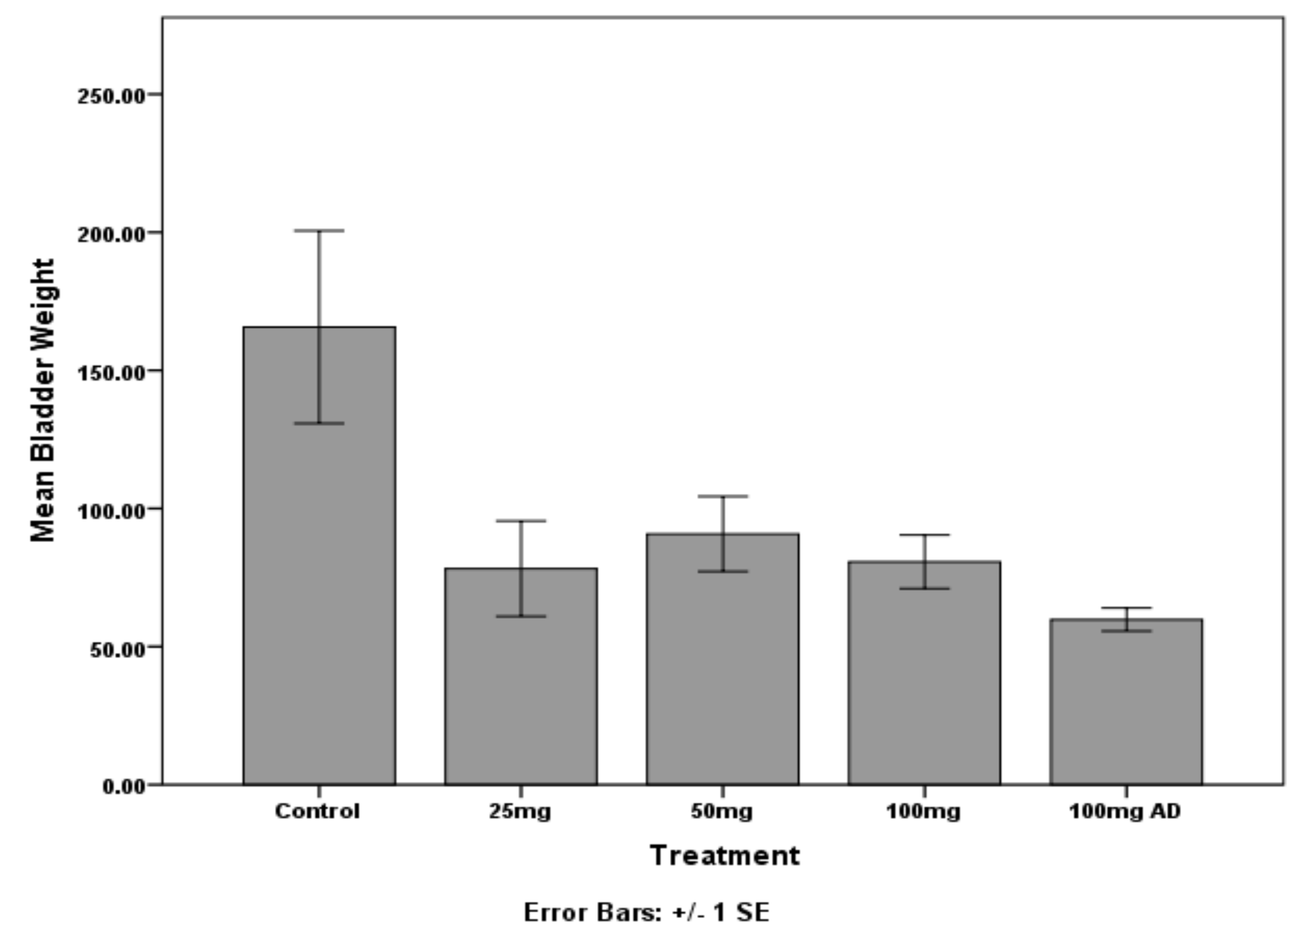
Mean Bladder Weight
Control
25mg
50mg
100mg
100mg AD
Treatment
Error Bars: +/- 1 SE

Figure 9. In the second BBN study, once daily IP CPX-POM doses of 25, 50, and 100 mg/kg for four weeks to 20 mice per treatment group resulted in significant differences ($p < 0.05$) in bladder weights versus controls. No difference observed in bladder weight between the CPX-POM once daily treatment groups nor between the (100 mg/kg CPX-POM) once daily versus three-times weekly treatment groups. As observed with the first BBN study, there was a moderate correlation between CPX-POM treatment and pathologic outcome that was not significant. Not shown, dose-dependent reductions in Ki67 and PCNA staining were observed. The minimum pharmacologic dose of CPX-POM in this mouse model of bladder cancer was not identified in this study.

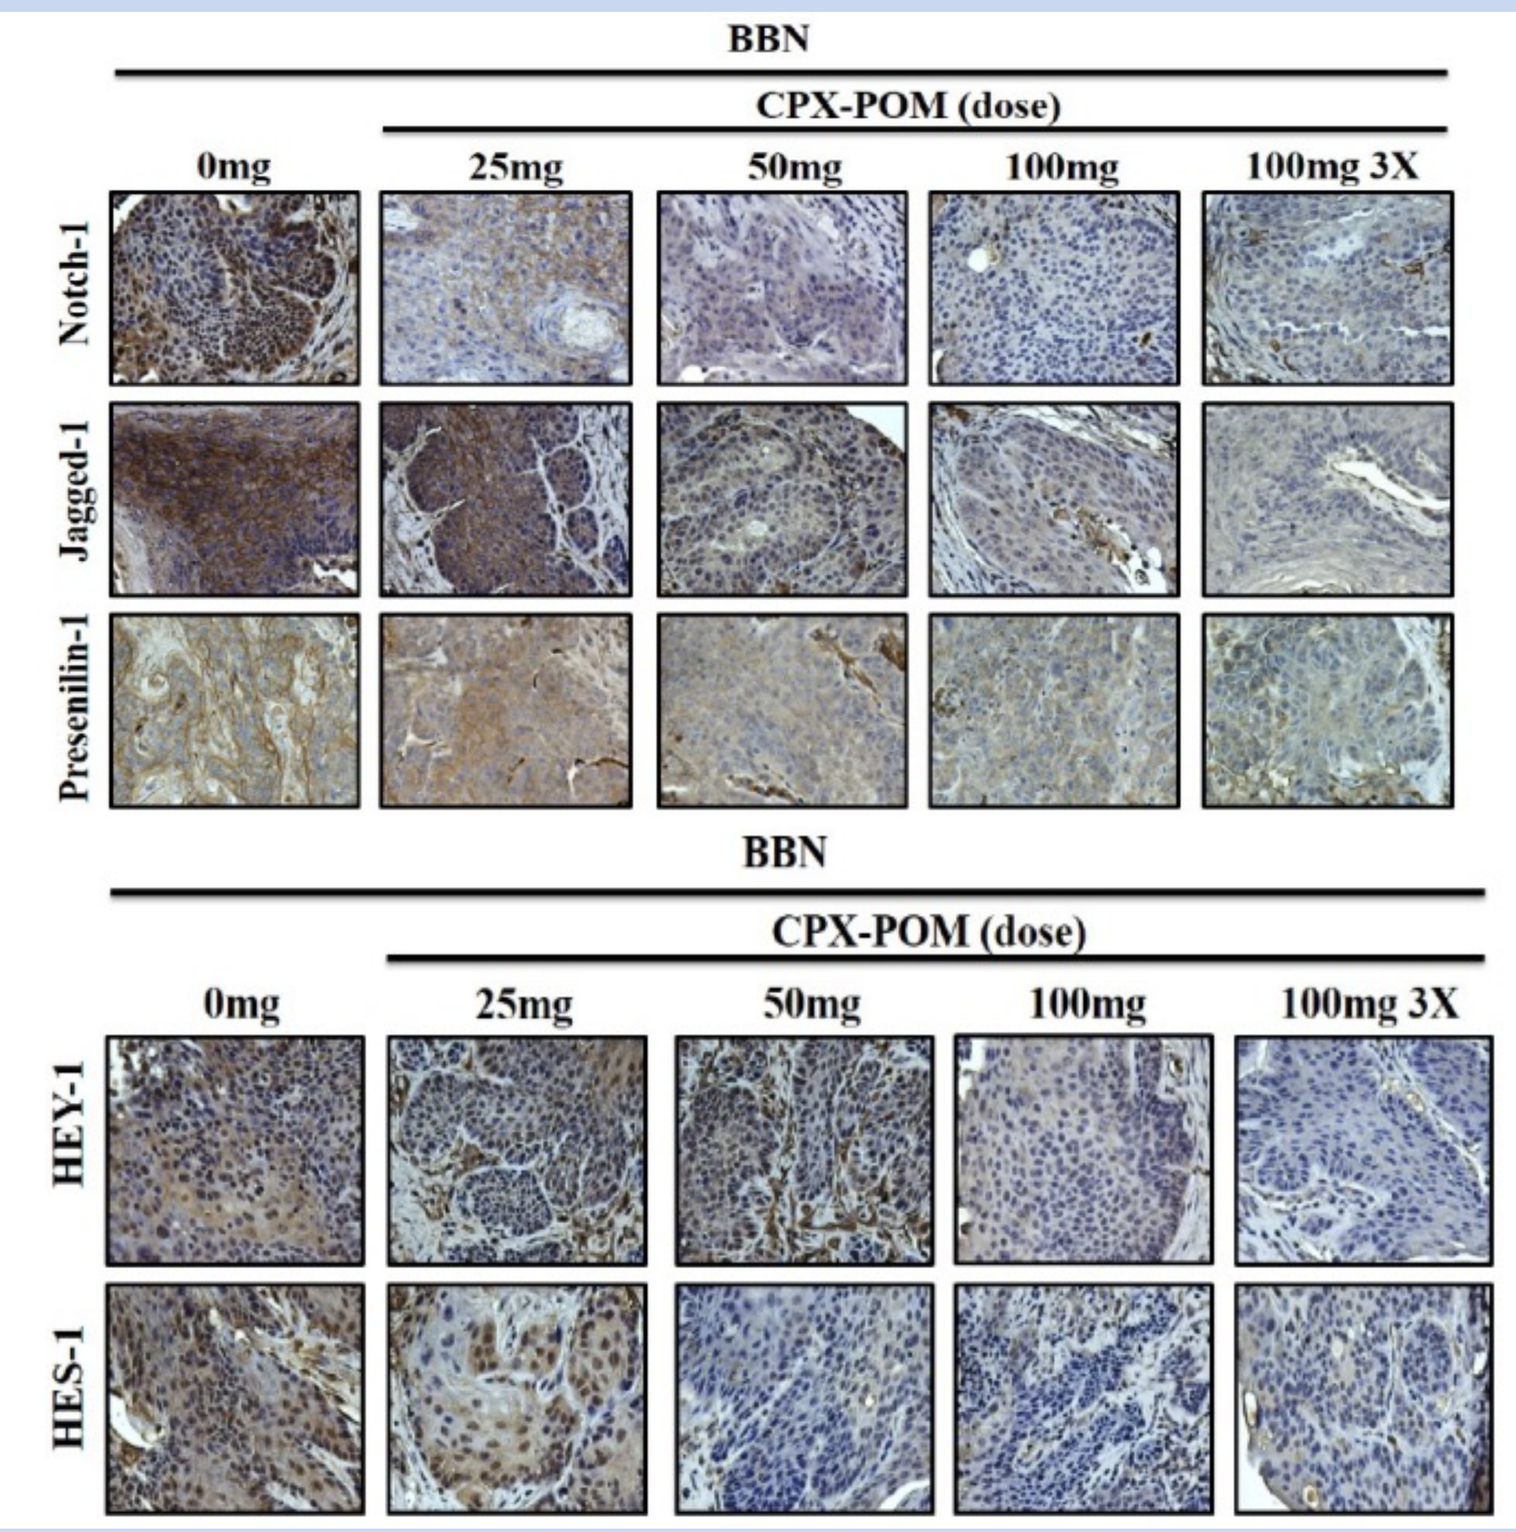
BBN
CPX-POM (dose)
0mg
25mg
50mg
100mg
100mg 3X
Notch-1
Jagged-1
Presenilin-1
HEY-1
HES-1

Figure 10. Immunohistochemical staining of bladder cancer tissues obtained from CPX-POM treated animals demonstrated reductions in five Notch signaling proteins including Notch 1, Jagged 1, Presenelin 1, Hes 1, and Hey 1 across the range of once daily CPX-POM doses as well as in animals receiving CPX-POM three-times weekly.

Ongoing and Future Studies

IND 132545 received FDA clearance to proceed with a first-in-human Phase 1 trial on 15 September 2017. The safety, dose tolerance, pharmacokinetics and pharmacodynamics of IV CPX-POM are currently being characterized in patients with advanced solid tumors at four US sites (NCT03348514) sponsored by CicloMed LLC. Development of a subcutaneous injectable formulation of CPX-POM is planned as a more convenient dosage form for self administration by bladder cancer patients.

Acknowledgements

Research reported in this presentation was supported by the National Cancer Institute Cancer Center Support Grant P30 CA168524 and used the CCSG Lead Development and Optimization Shared Resource. Several studies described herein were funded by a public-private partnership between The Institute for Advancing Medical Innovation at the University of Kansas Medical Center and CicloMed LLC, Kansas City, MO.

[1]University of Kansas Medical Center, Kansas City, KS; [2]University of Kansas, Lawrence, KS; [3]Navigator LSA, Wilmington, NC; [4]Alba BioPharm Advisors Inc., Durham, NC