Fosciclopirox Suppresses Growth of High-Grade Urothelial Cancer by Targeting Notch Signaling

Scott J. Weir[1,2,3,4], Prasad Dandewate[1], Prabhu Ramamoorthy[1] , Partha Ranjaran[1,], Robyn Wood[3], Amanda Brinker[1,3] , Ben Woolbright[5], Mehmet Tanol[7], Tammy Ham[8], William McCulloch[9], Michael Dalton[10], Michael J. Baltezor[3,6], Roy A. Jensen[1,,11], John A. Taylor, III[1,5], Shrikant Anant[1,4]

Abstract #
6405

Poster Board #
29

Fosciclopirox

Ciclopirox (CPX) is an FDA-approved topical antifungal agent that has demonstrated preclinical anticancer activity in solid and hematologic malignancies. It's clinical utility as an oral anticancer agent, however, is limited by poor oral bioavailability and gastrointestinal toxicity.

Fosciclopirox, the phosphorylmethoxy ester of ciclopirox (Ciclopirox Prodrug, CPX-POM), is rapidly and completely metabolized to CPX, the active metabolite, which subsequently undergoes renal elimination resulting in urine concentrations of CPX that exceed *in vitro* IC50's > 27-fold in humans at well tolerated doses.

Fosciclopirox is being developed for the treatment of urothelial cancers through a public-private partnership between the Institute for Advancing Medical Innovation at the University of Kansas Medical Center and CicloMed LLC. The safety, dose tolerance, pharmacokinetics and pharmacodynamics of CPX-POM were characterized in 19 patients with advanced solid tumors participating in a US multicenter, First-in-Human Phase 1, open-label, dose escalation study (NCT03348514). An expansion cohort study in 12 cisplatin-ineligible muscle invasive bladder cancer (MIBC) patients receiving two 21-day treatment cycles of CPX-POM prior to radical cystectomy (RC) is underway. Evidence of pharmacologic activity is being characterized in bladder tumor tissues obtained at RC.

Bladder Cancer

Bladder cancer (BCa) is a devastating disease that currently ranks as the fourth most common cancer among men and the sixth most common among men and women combined. The American Cancer Society estimates that in 2020 alone, 81,400 new cases will be diagnosed in the U.S. and 17,980 will die of the disease. Bladder cancer is defined as two diseases, non-muscle invasive (NMIBC) and muscle invasive (MIBC) bladder cancer, each with different treatment approaches and outcomes. A 2014 survey estimated that in the US alone, there were a total of 696,400 people living with BCa. In spite of having the highest recurrence rate of all known malignancies, as well as high rates of progression, the overall 5-year survival rate for BCa is 77%. Due to the high rates of recurrence and progression which requires life-long surveillance, BCa is the most expensive cancer to treat on a per-patient-per-lifetime basis.

Conclusions

In vitro and *in vivo* preclinical studies suggest CPX acts at least in part by engaging cancer stem cell pathways in high-grade urothelial cancer models. Following administration of CPX-POM to mice with chemically-induced *carcinoma in situ*, engagement of the active metabolite with Notch 1 is associated with lower tumor size, migration to lower stage tumors, inhibition of cell proliferation and reductions in Notch 1, Presenilin 1, and Hey 1 in bladder tumor tissues. Molecular docking and CETSA studies demonstrate CPX binds with γ-secretase complex proteins Presenilin 1 and Nicastrin. Higher levels of Presenilin 1 and Nicastrin Iin bladder tumor tissues are associated with patient survival.

Acknowledgements

We thank Dr. Michael J. McKenna, who inspired us to continue this work even as he fought a losing battle to cancer. This drug development program has and continues to utilize Lead Development and Optimization as well as Clinical Pharmacology shared resources supported by the National Cancer Institute Cancer Center Support Grant P30 CA168524.

CPX Inhibits Proliferation and Colony Formation

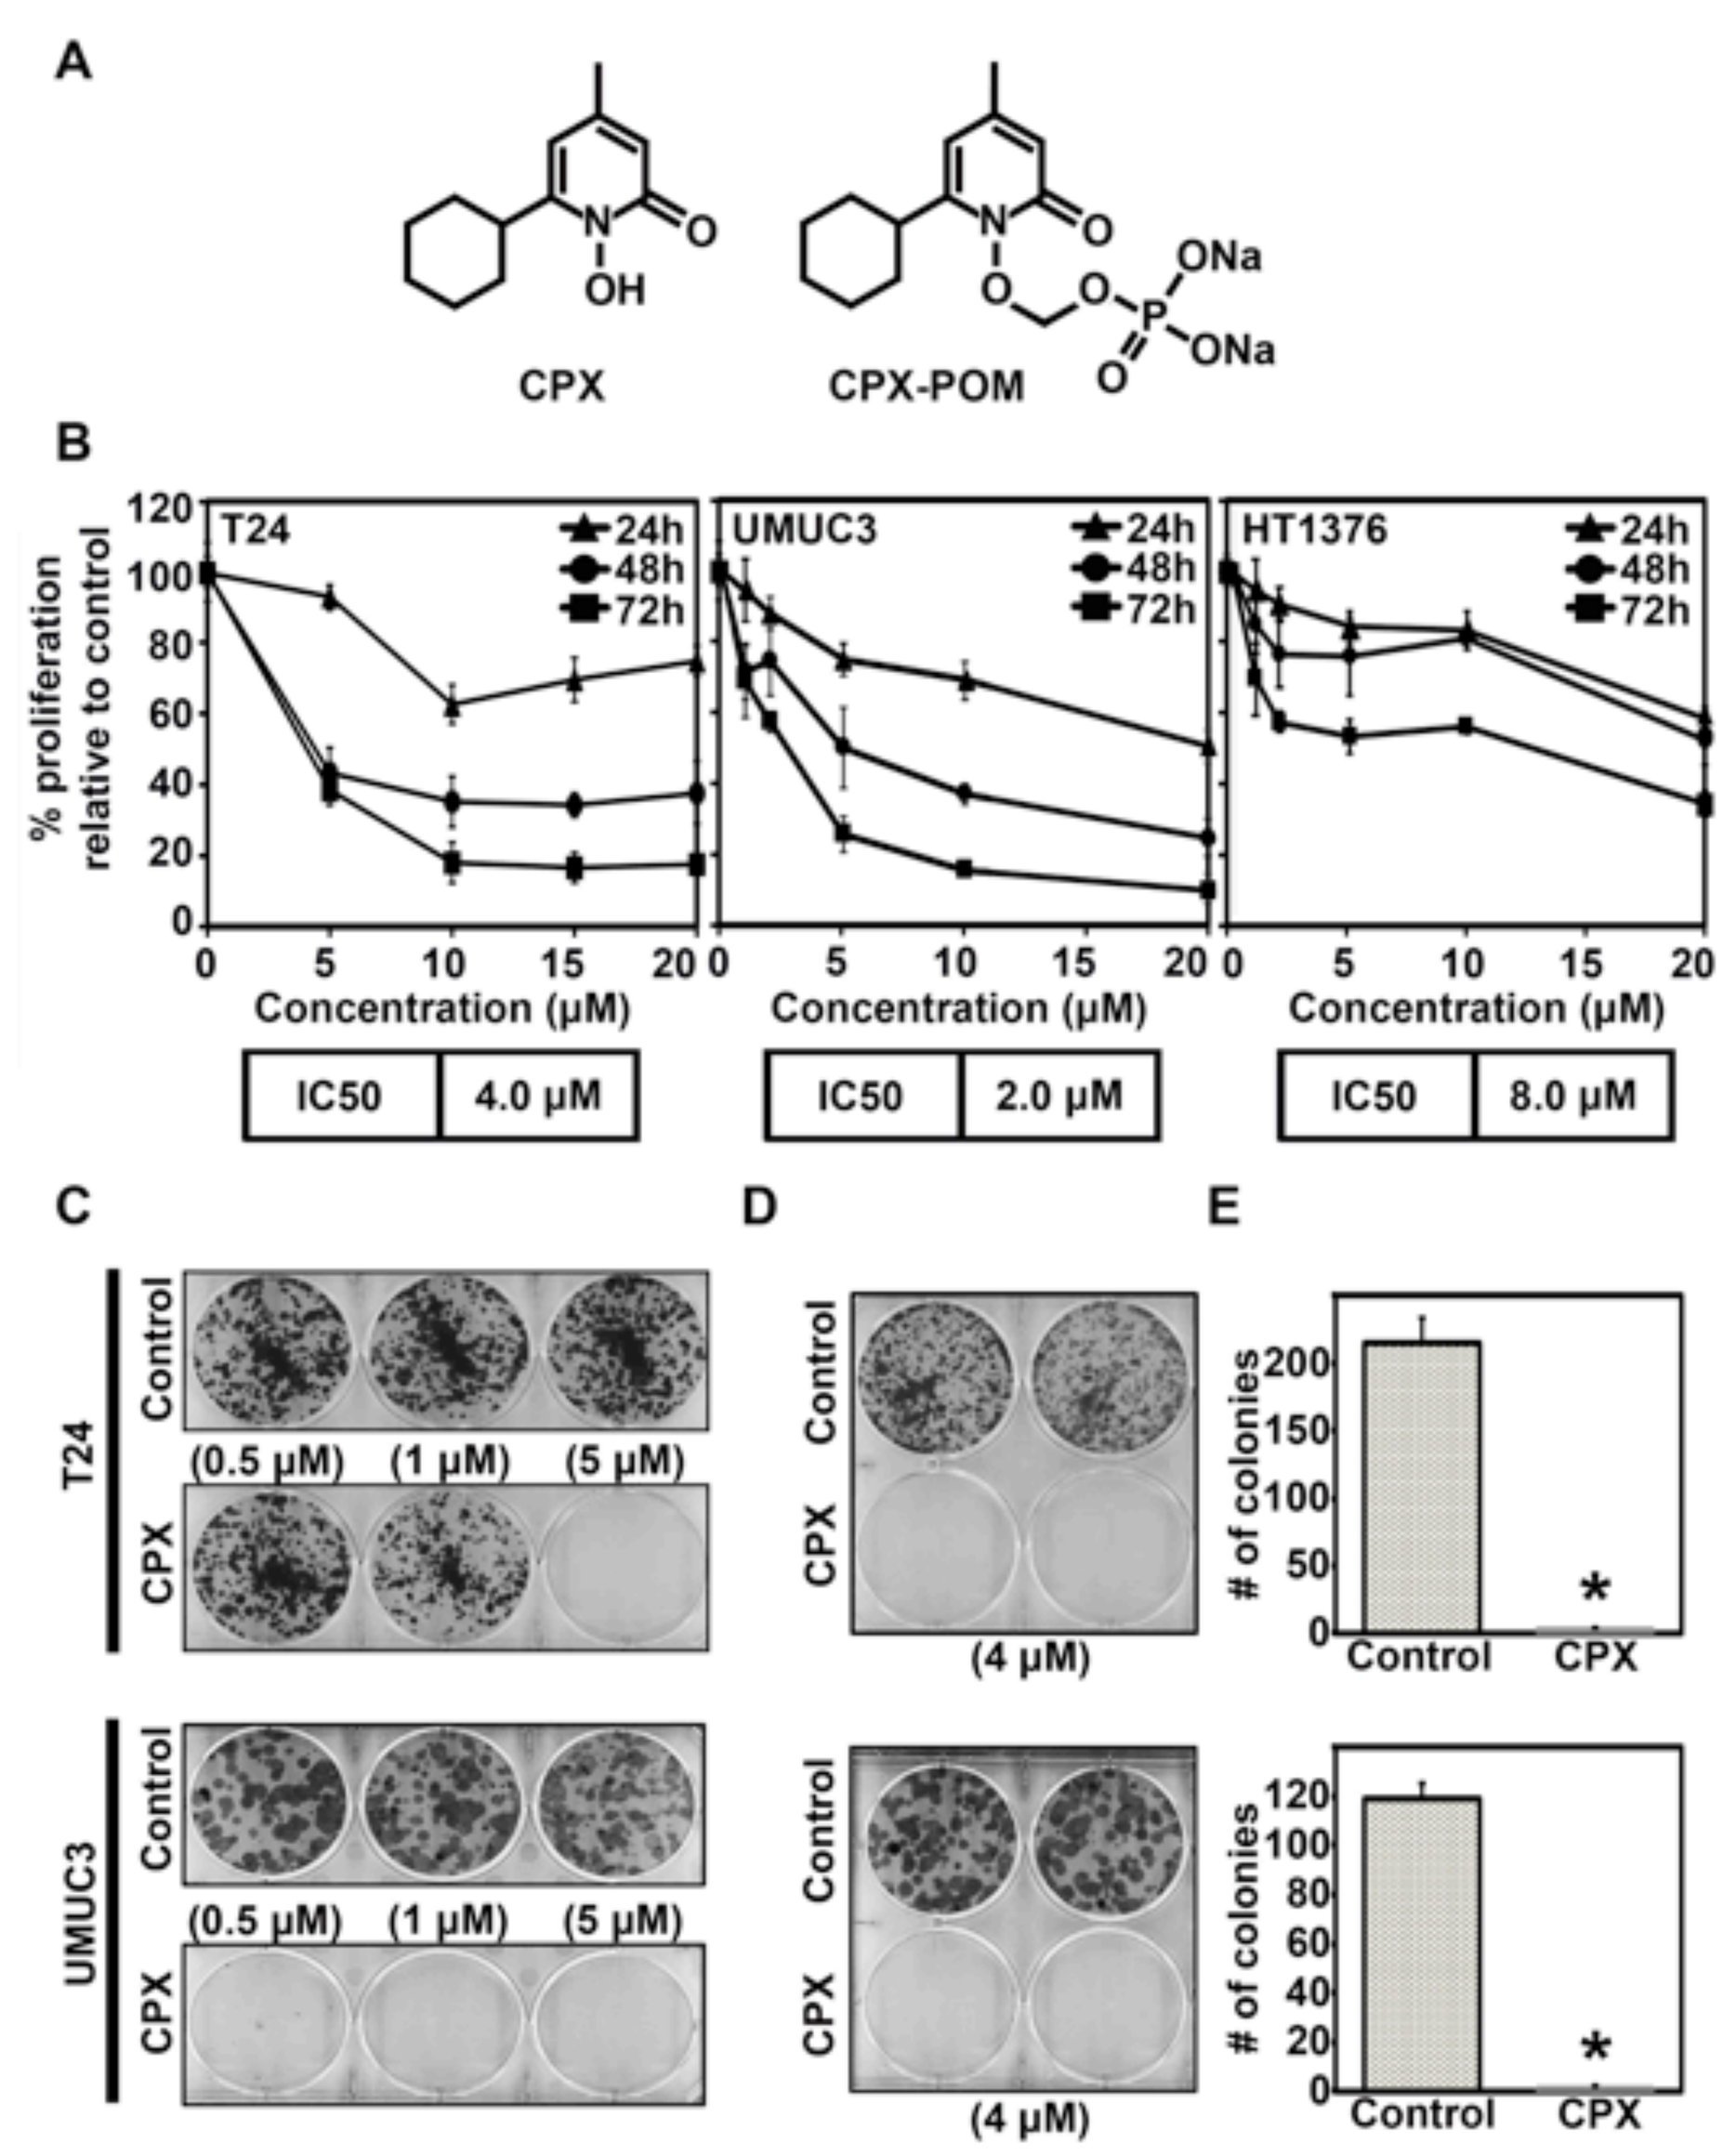
A
CPX
CPX-POM
B
T24
UMUC3
HT1376
24h
48h
72h
% proliferation relative to control
Concentration (μM)
IC50 4.0 μM
IC50 2.0 μM
IC50 8.0 μM
C
D
E
T24
UMUC3
Control
CPX
(0.5 μM) (1 μM) (5 μM)
(4 μM)
of colonies

Figure 1. A. Structures of Fosciclopirox (CPX-POM) and its active metabolite ciclopirox (CPX). **B.** Exposure to CPX significantly suppressed proliferation of T24, UMUC3, and HT1376 cells in a concentration- and time-dependent manner. IC_{50} values for CPX were determined to be 2μM, 4μM, and 8μM at 72 hours exposure for T24, UMUC-3, and HT1376 cell lines, respectively. **C.** Treatment with CPX significantly suppressed the number of colonies in both T24 and UMUC3 cells. **D & E.** Colony formation was significantly suppressed when the respective cell lines were exposed to CPX at IC_{50} concentrations.

CPX Induces Cell Cycle Arrest and Apoptosis

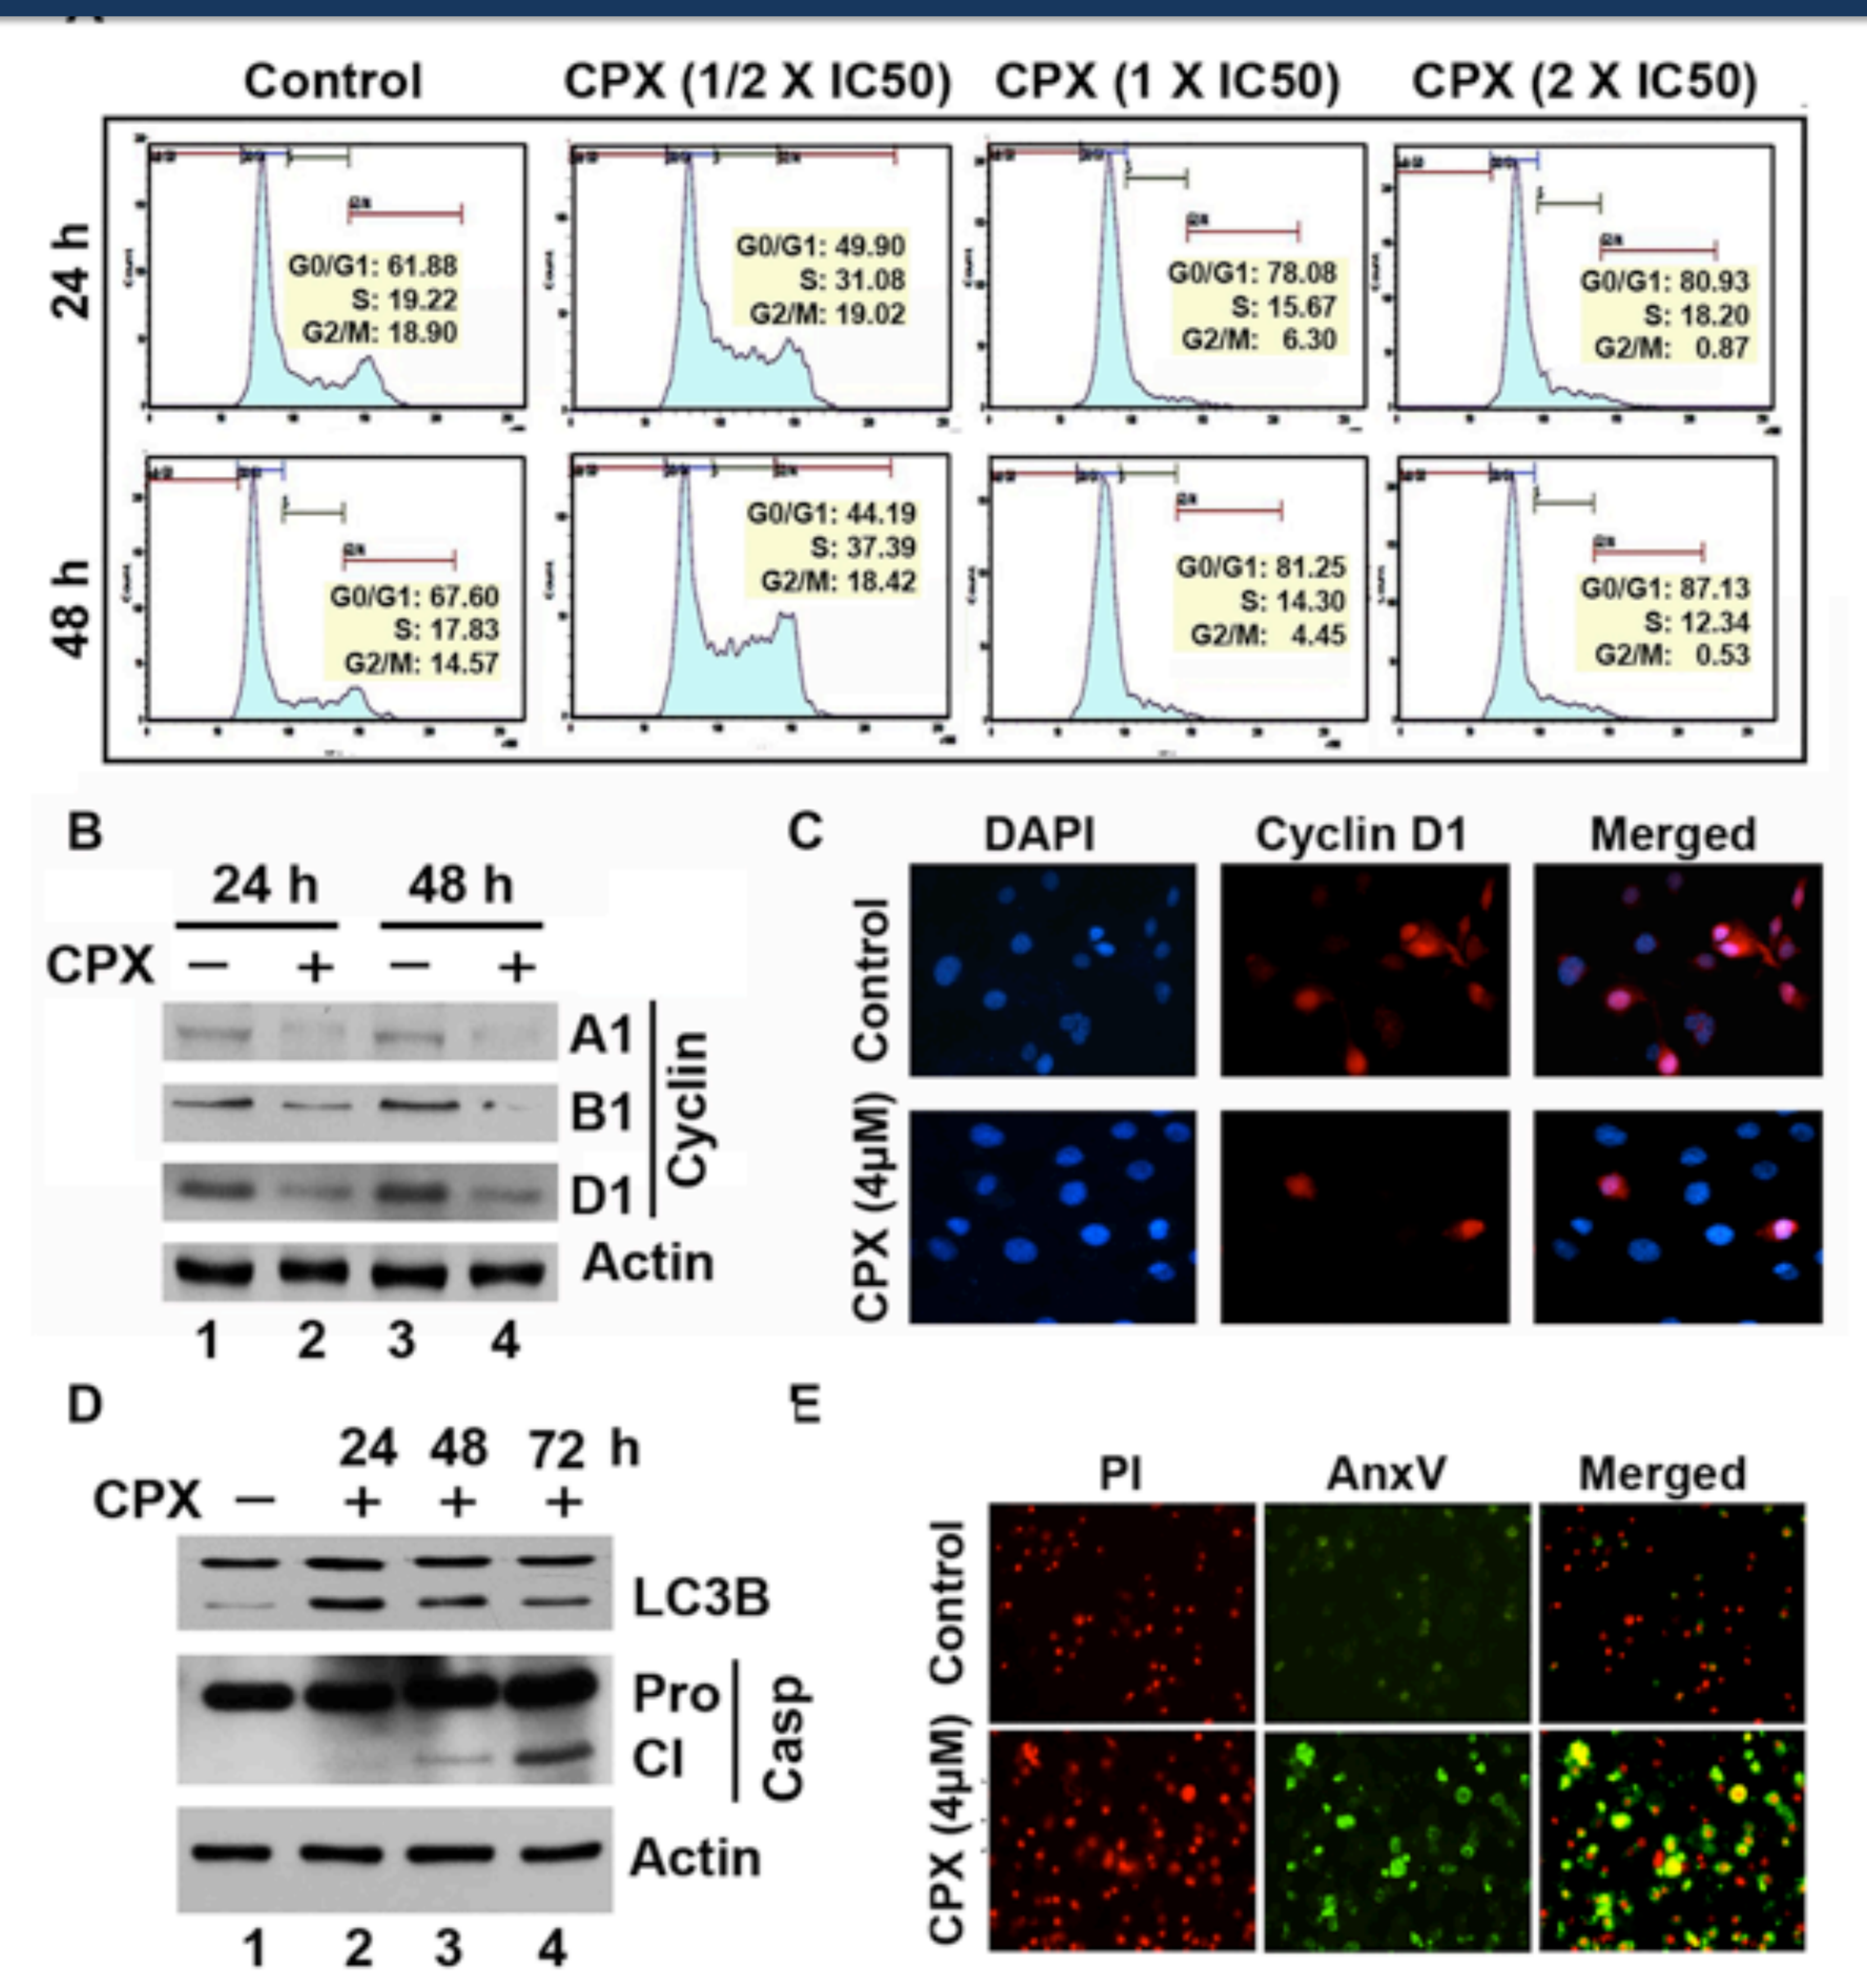
Control
CPX (1/2 X IC50)
CPX (1 X IC50)
CPX (2 X IC50)
24 h
48 h
G0/G1: 61.88 S: 19.22 G2/M: 18.90
G0/G1: 49.90 S: 31.08 G2/M: 19.02
G0/G1: 78.08 S: 15.67 G2/M: 6.30
G0/G1: 80.93 S: 18.20 G2/M: 0.87
G0/G1: 67.60 S: 17.83 G2/M: 14.57
G0/G1: 44.19 S: 37.39 G2/M: 18.42
G0/G1: 81.25 S: 14.30 G2/M: 4.45
G0/G1: 87.13 S: 12.34 G2/M: 0.53
B
CPX
A1
B1
D1
Cyclin
Actin
C
DAPI
Cyclin D1
Merged
CPX (4μM)
D
LC3B
Pro
Cl
Casp
E
PI
AnxV

Figure 2. A. Treatment with ½ IC_{50} resulted in a significant increase in cells in S phase while increased concentrations of CPX resulted in G0/G1 arrest. **B.** In T24 cells, cyclin A1, B1 and D1 were decreased following incubation with CPX at the IC_{50} value of 4μM. **C.** Given cyclin D1 plays a significant role in the progression from G1 to S phase of the cell cycle, we confirmed its expression was reduced by immunofluorescence. **D.** At 4μM CPX, key effector proteins for autophagy and apoptosis, LC3B and cleaved caspase 3, respectively, were increased. **E.** CPX effects on apoptosis were further confirmed by immunofluorescence for annexin V. Taken together, these data suggest at early time points, CPX induces cells to undergo autophagy which switches to apoptosis.

CPX Inhibits Cell Migration, Invasion and Spheroid Growth

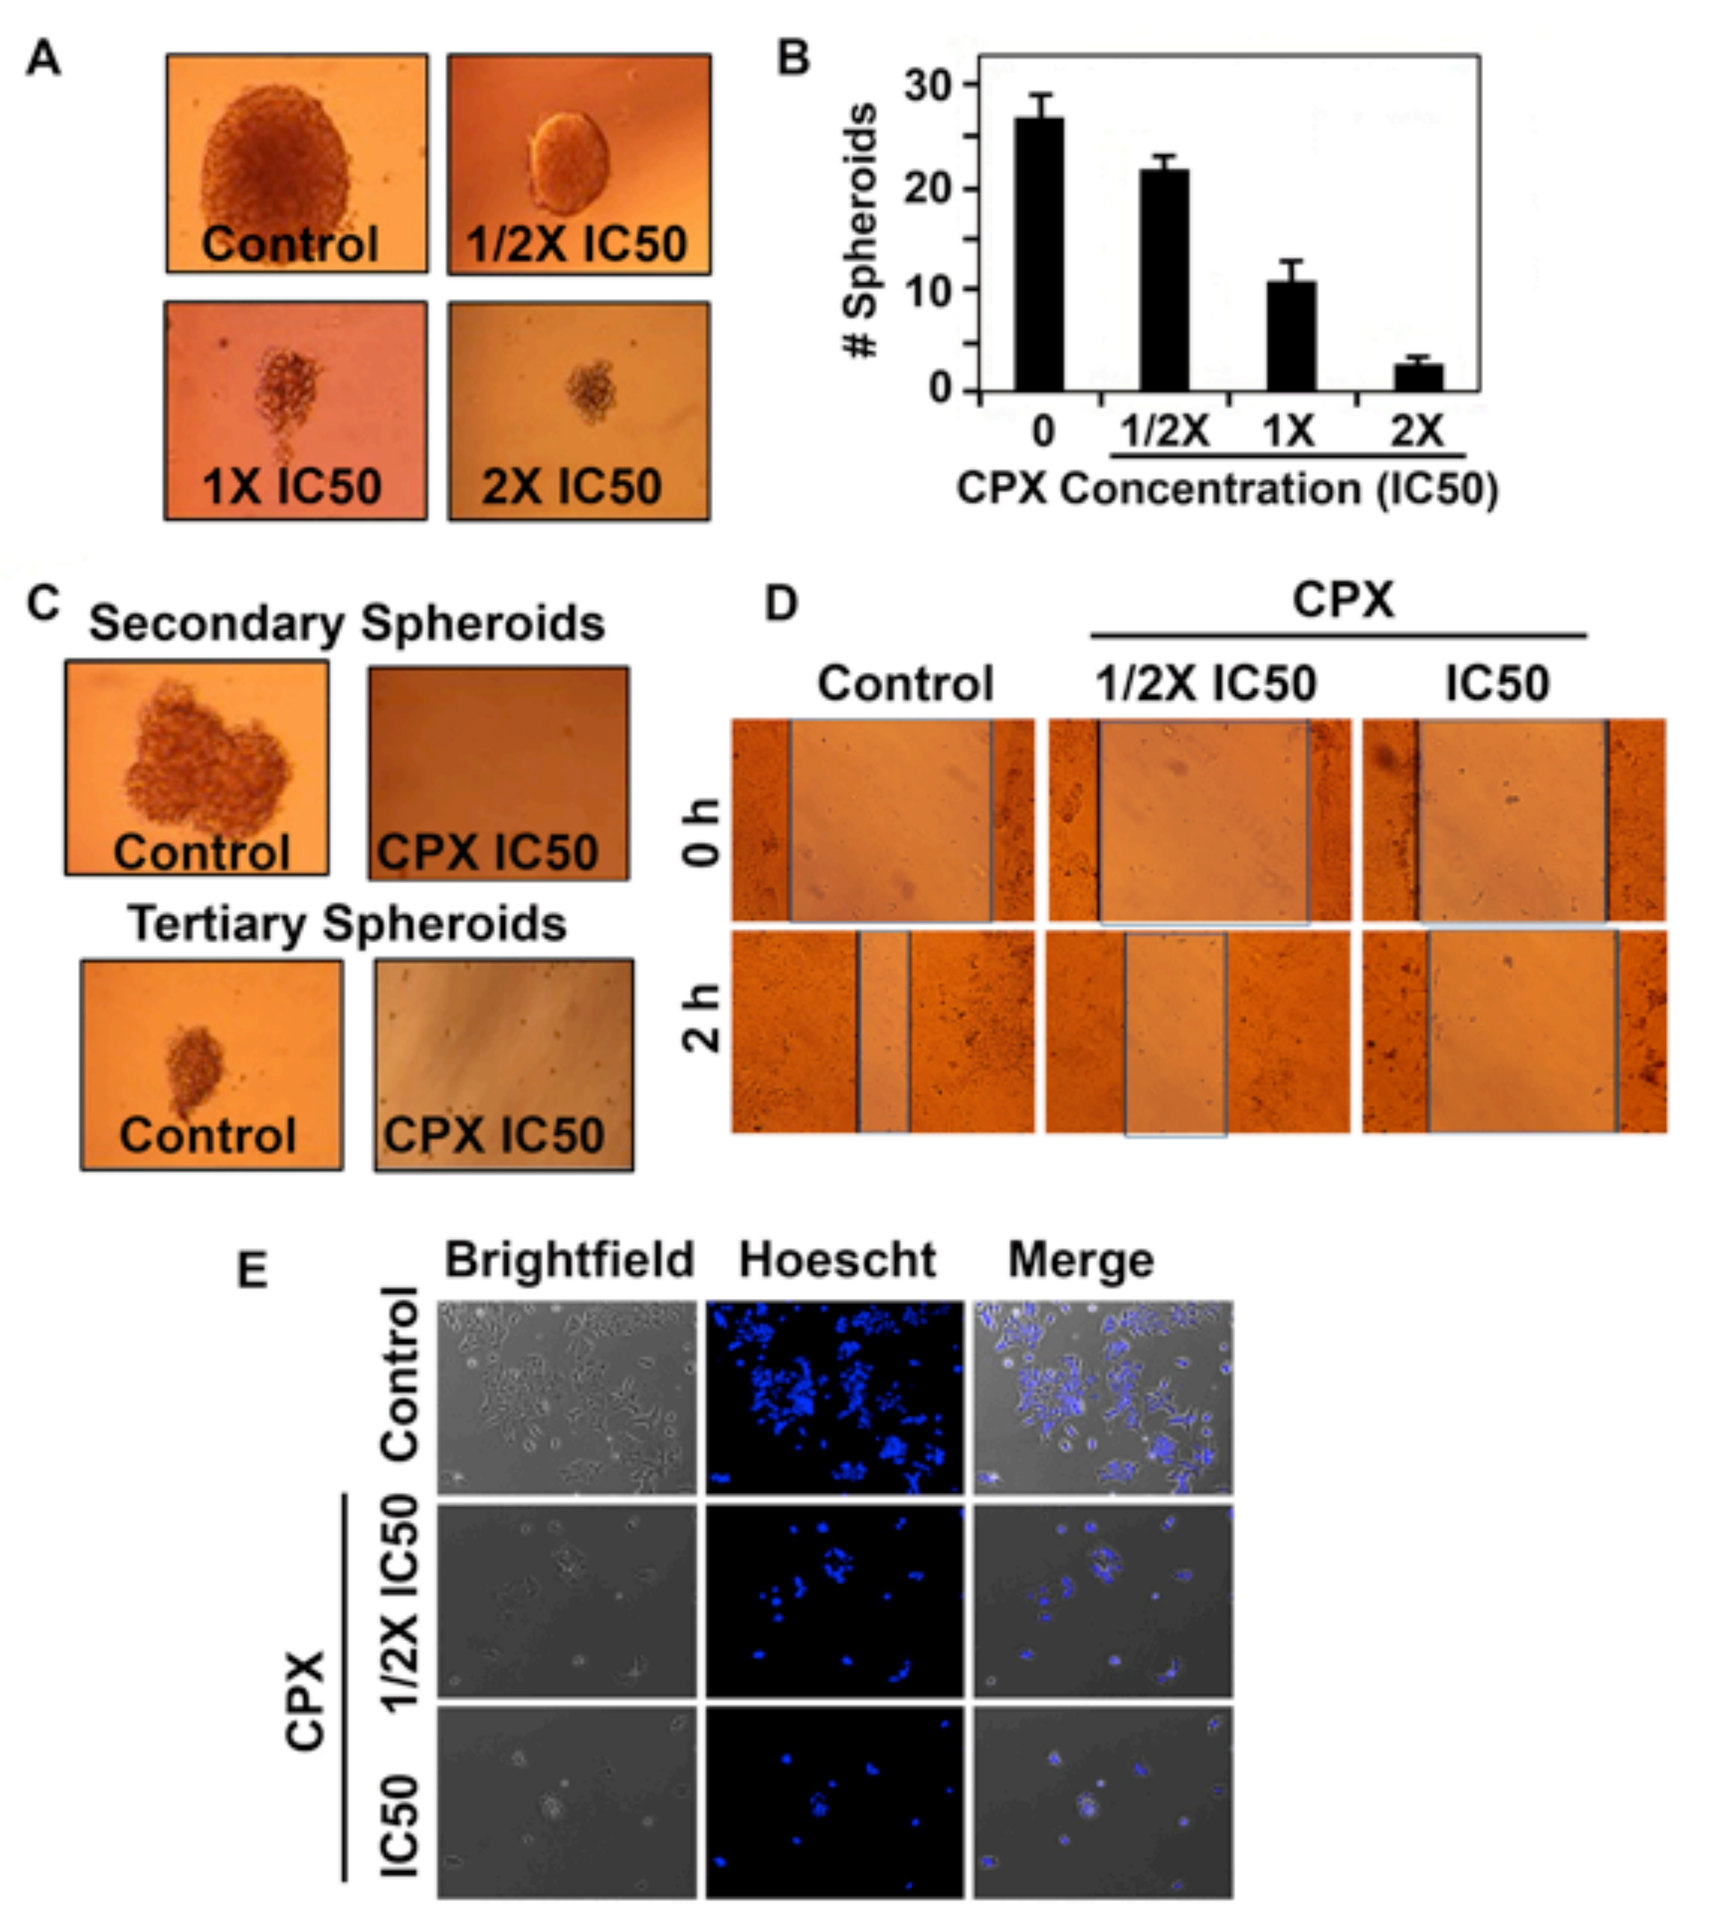
A
Control
1/2X IC50
1X IC50
2X IC50
B
Spheroids
CPX Concentration (IC50)
C
Secondary Spheroids
Tertiary Spheroids
CPX IC50
D
CPX
0 h
2 h
E
Brightfield
Hoescht
Merge

Figure 3. A & B. T24 cells were plated in ultra-low attachment plates under conditions that favor growth of spheroids. CPX exposure resulted in significant reductions in size and number of spheroids in a concentration-dependent manner. **C.** No spheroids were formed following replating of primary spheroids exposed to CPX suggesting CPX reduced the viability of cancer stem cells. **D.** In the wound healing assay, CPX significantly reduced closure of the cell-free area indicating the active metabolite decreased cell migration. **E.** CPX at ½ IC_{50} suppressed invasion of T24 cells through Matrigel.

CPX Inhibits Notch Signaling through Inactivation of the γ-Secretase Complex

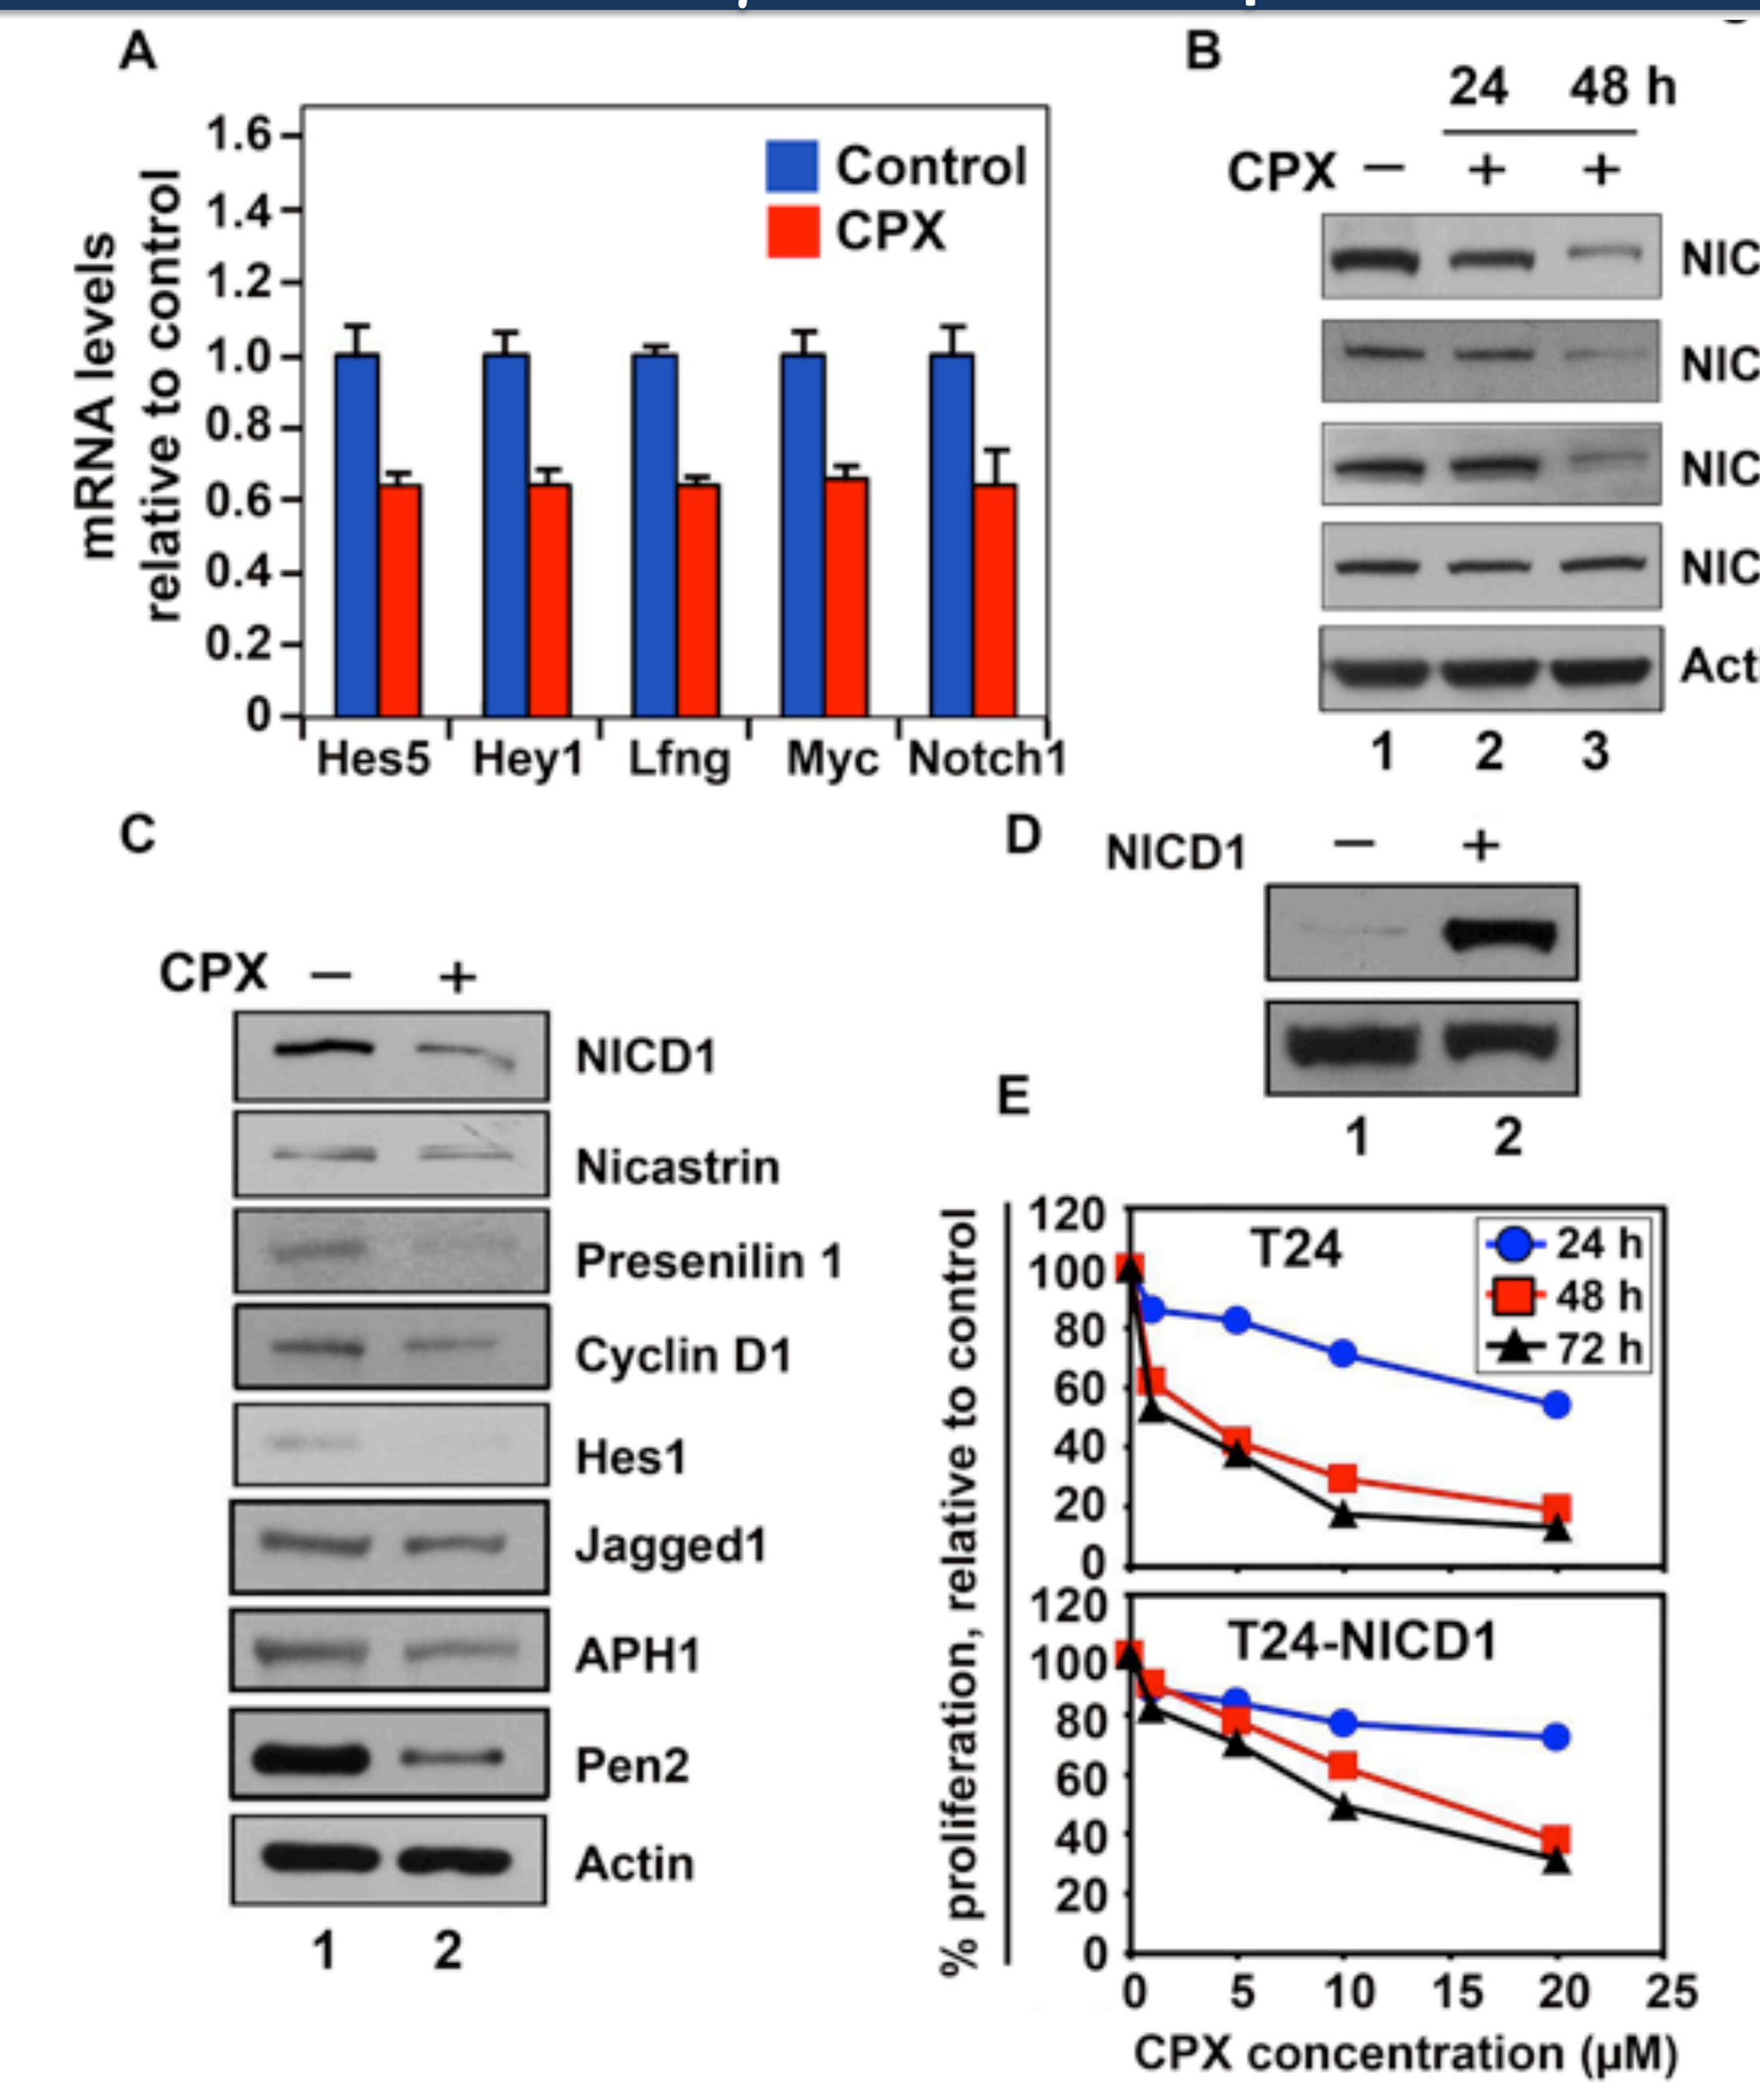
A
mRNA levels relative to control
Control
CPX
Hes5
Hey1
Lfng
Myc
Notch1
B
24 48 h
NICD1
NICD2
NICD3
NICD4
Actin
C
Nicastrin
Presenilin 1
Cyclin D1
Hes1
Jagged1
APH1
Pen2
D
E
T24
T24-NICD1
24 h
48 h
72 h
% proliferation, relative to control
CPX concentration (μM)

Figure 4. A. CPX at the IC_{50} decreased Notch signaling-related proteins in T24 cells. **B.** Western blot analysis demonstrated CPX at ½ IC_{50} suppressed activation of Notch 1, Notch 2, and Notch 3. **C.** CPX reduced expression of Jagged1, Hes-1 and Cyclin D1. Expression of γ-secretase proteins Presenilin 1, Nicastrin, APH-1, and PEN-2. **D.** The Notch intracellular domain was ectopically expressed to determine the significance of Notch inhibition on cell proliferation and stemness. **E.** Ectopic overexpression of the Notch intracellular domain (NICD) partially reversed the effects of CPX on T24 cell proliferation. Together, these data suggest CPX inhibits bladder cancer growth *in vitro,* at least in part, by suppressing the Notch 1 signaling pathway.

Fosciclopirox Suppresses Bladder Tumorigenesis in a Chemically-Induced Model

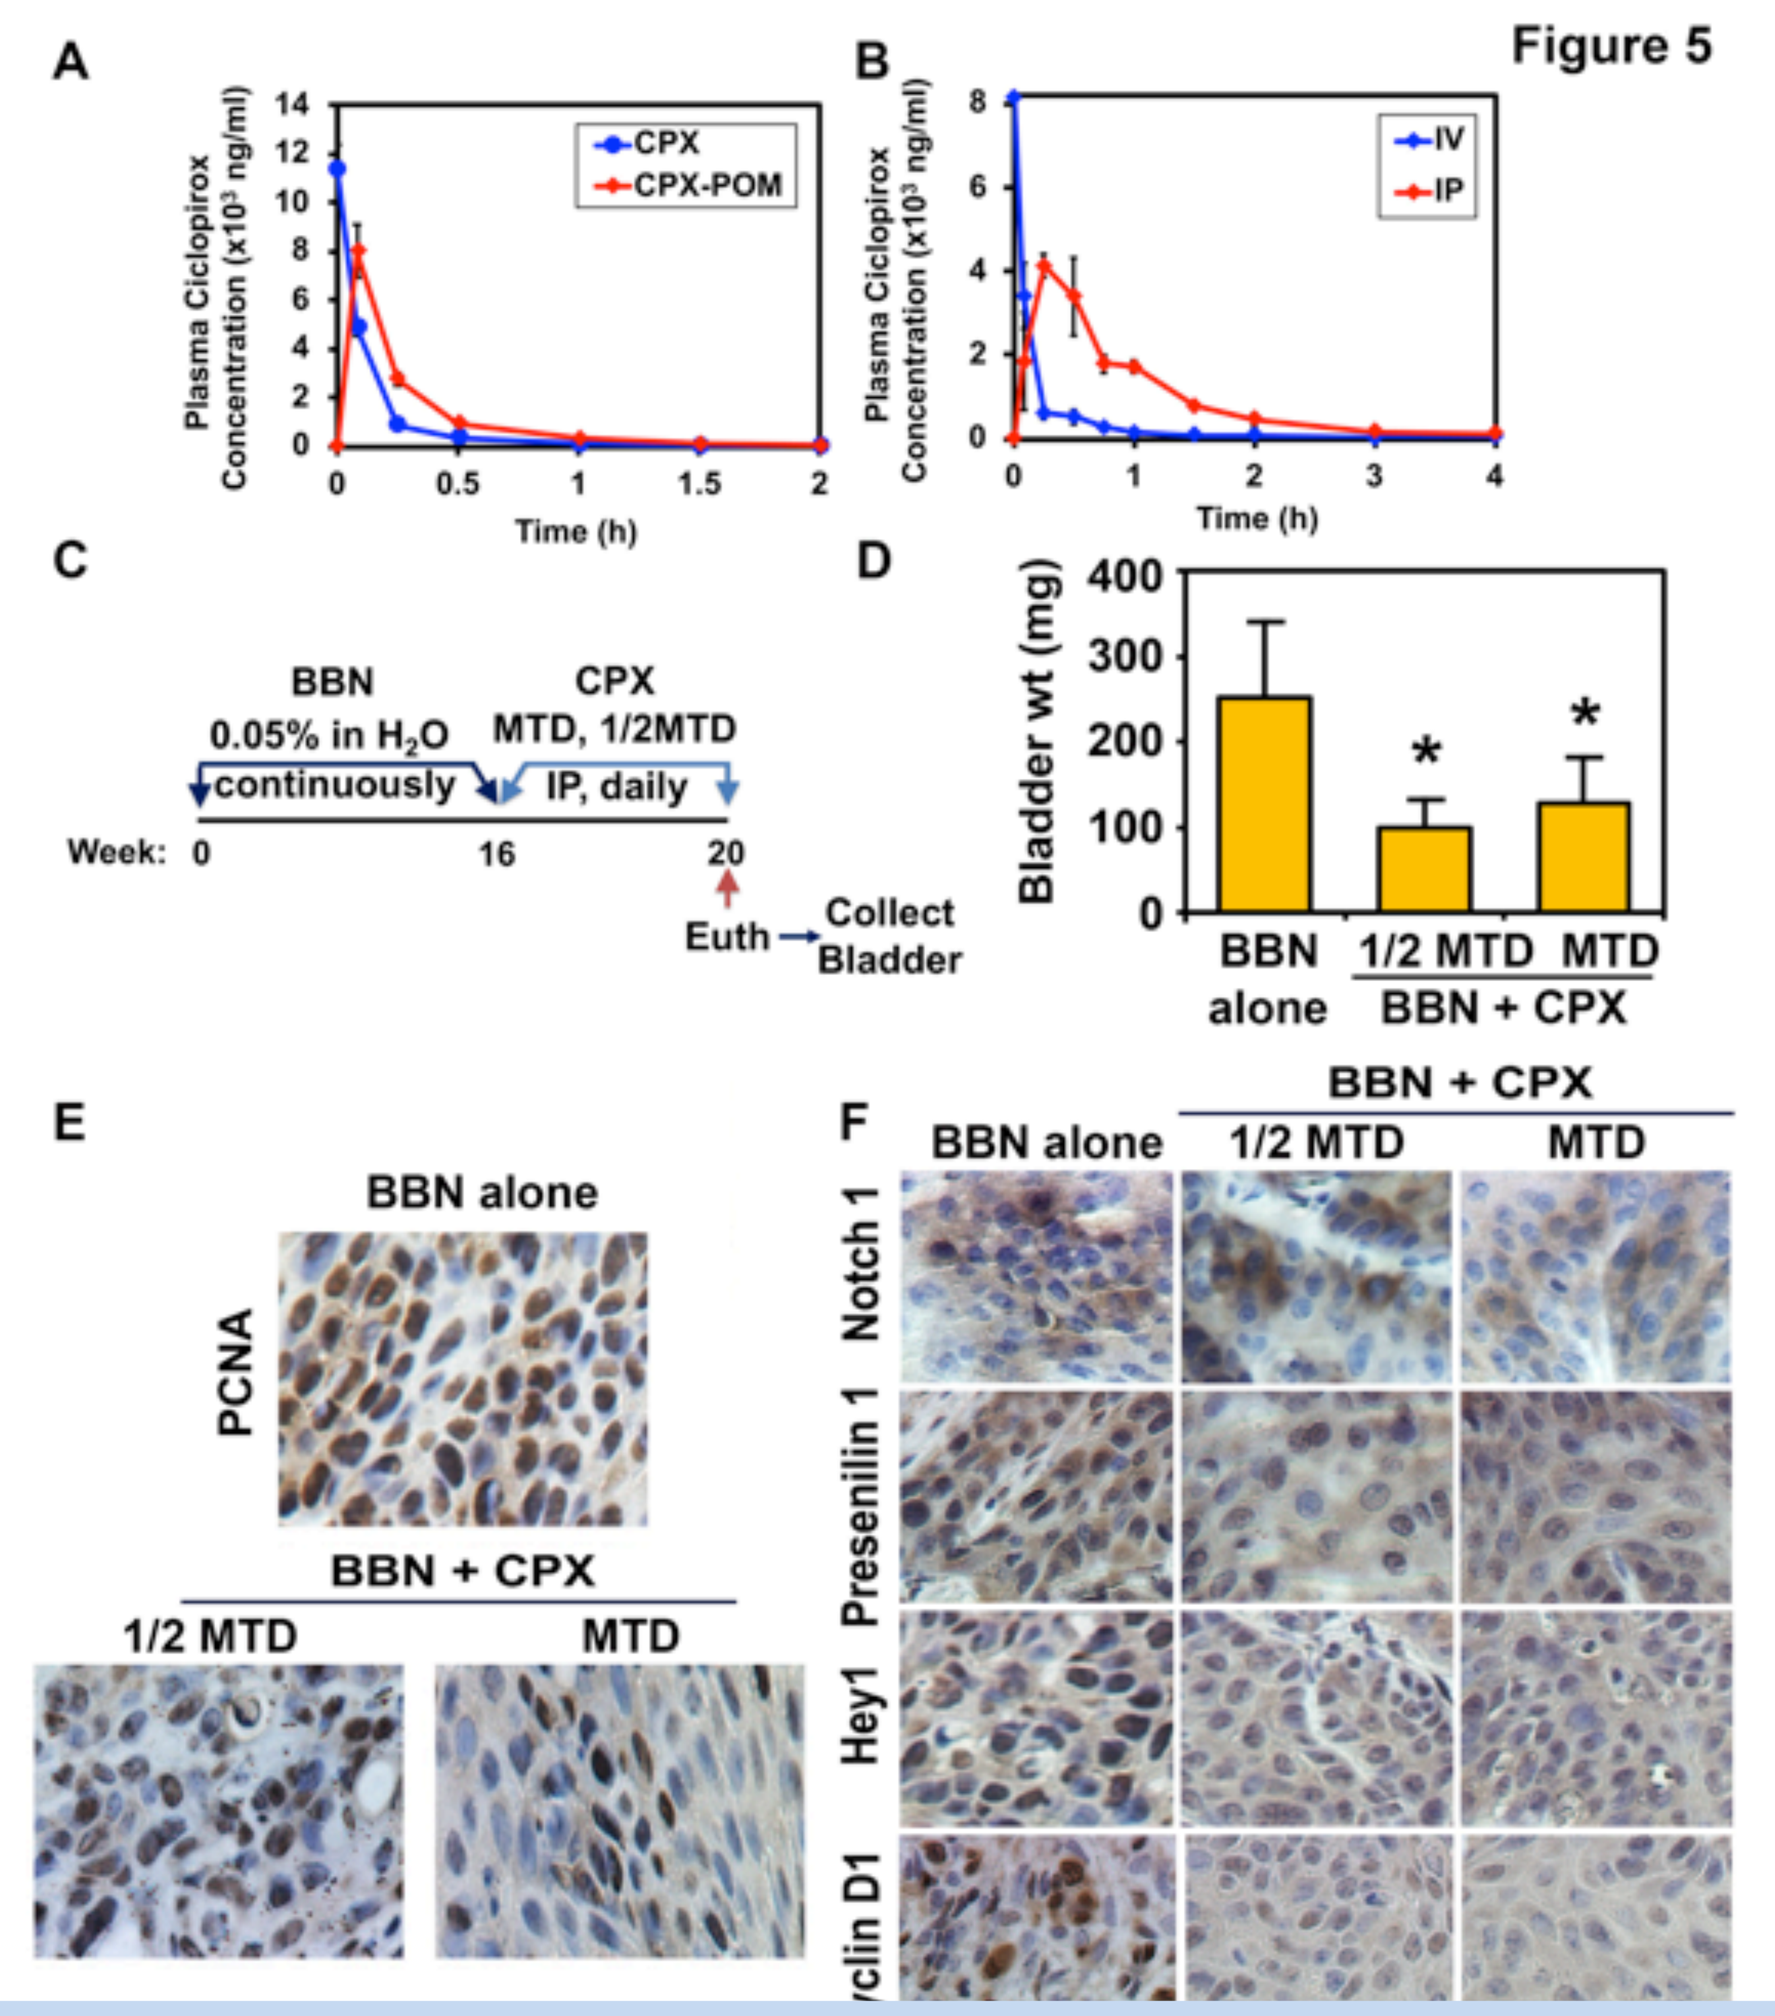
Figure 5
A
B
Plasma Ciclopirox Concentration (x10³ ng/ml)
CPX
CPX-POM
IV
IP
Time (h)
C
BBN
0.05% in H₂O
continuously
CPX
MTD, 1/2MTD
IP, daily
Week: 0 16 20
Euth
Collect Bladder
D
Bladder wt (mg)
BBN alone
1/2 MTD
MTD
BBN + CPX
E
PCNA
F
Notch 1
Presenilin 1
Hey1

Figure 4. A. Administration of CPX-POM and the olamine salt of CPX IV in mice results in similar systemic exposure of the active metabolite demonstrating complete metabolism of the prodrug. CPX-POM was not detected in plasma. **B.** The absolute bioavailability of CPX following IV and IP administration of CPX-POM in mice was 60%. Not shown, IP CPX-POM at ¼MTD, ½ IC_{50} MTD and MTD resulted in steady-state urine CPX concentrations 15-30 fold greater than the ½ IC_{50} **C.** The BBN mouse model of bladder cancer was employed to determine preclinical proof of principle for CPX-POM. To establish *carcinoma in situ*, mice were given 0.05% BBN in drinking water *ad libitum* for 16 weeks. **D.** Bladder weights in CPX-POM treated animals were significantly less than vehicle controls. There were no significant differences in bladder weights between the two CPX-POM treatment groups. **E & F.** Reduction in PCNA staining was observed in bladder tumor tissues in CPX-POM treated animals. IHC analysis demonstrated decreased staining for Notch 1, Presenilin 1, Hey 1, and Cyclin D1 in a dose-dependent fashion.

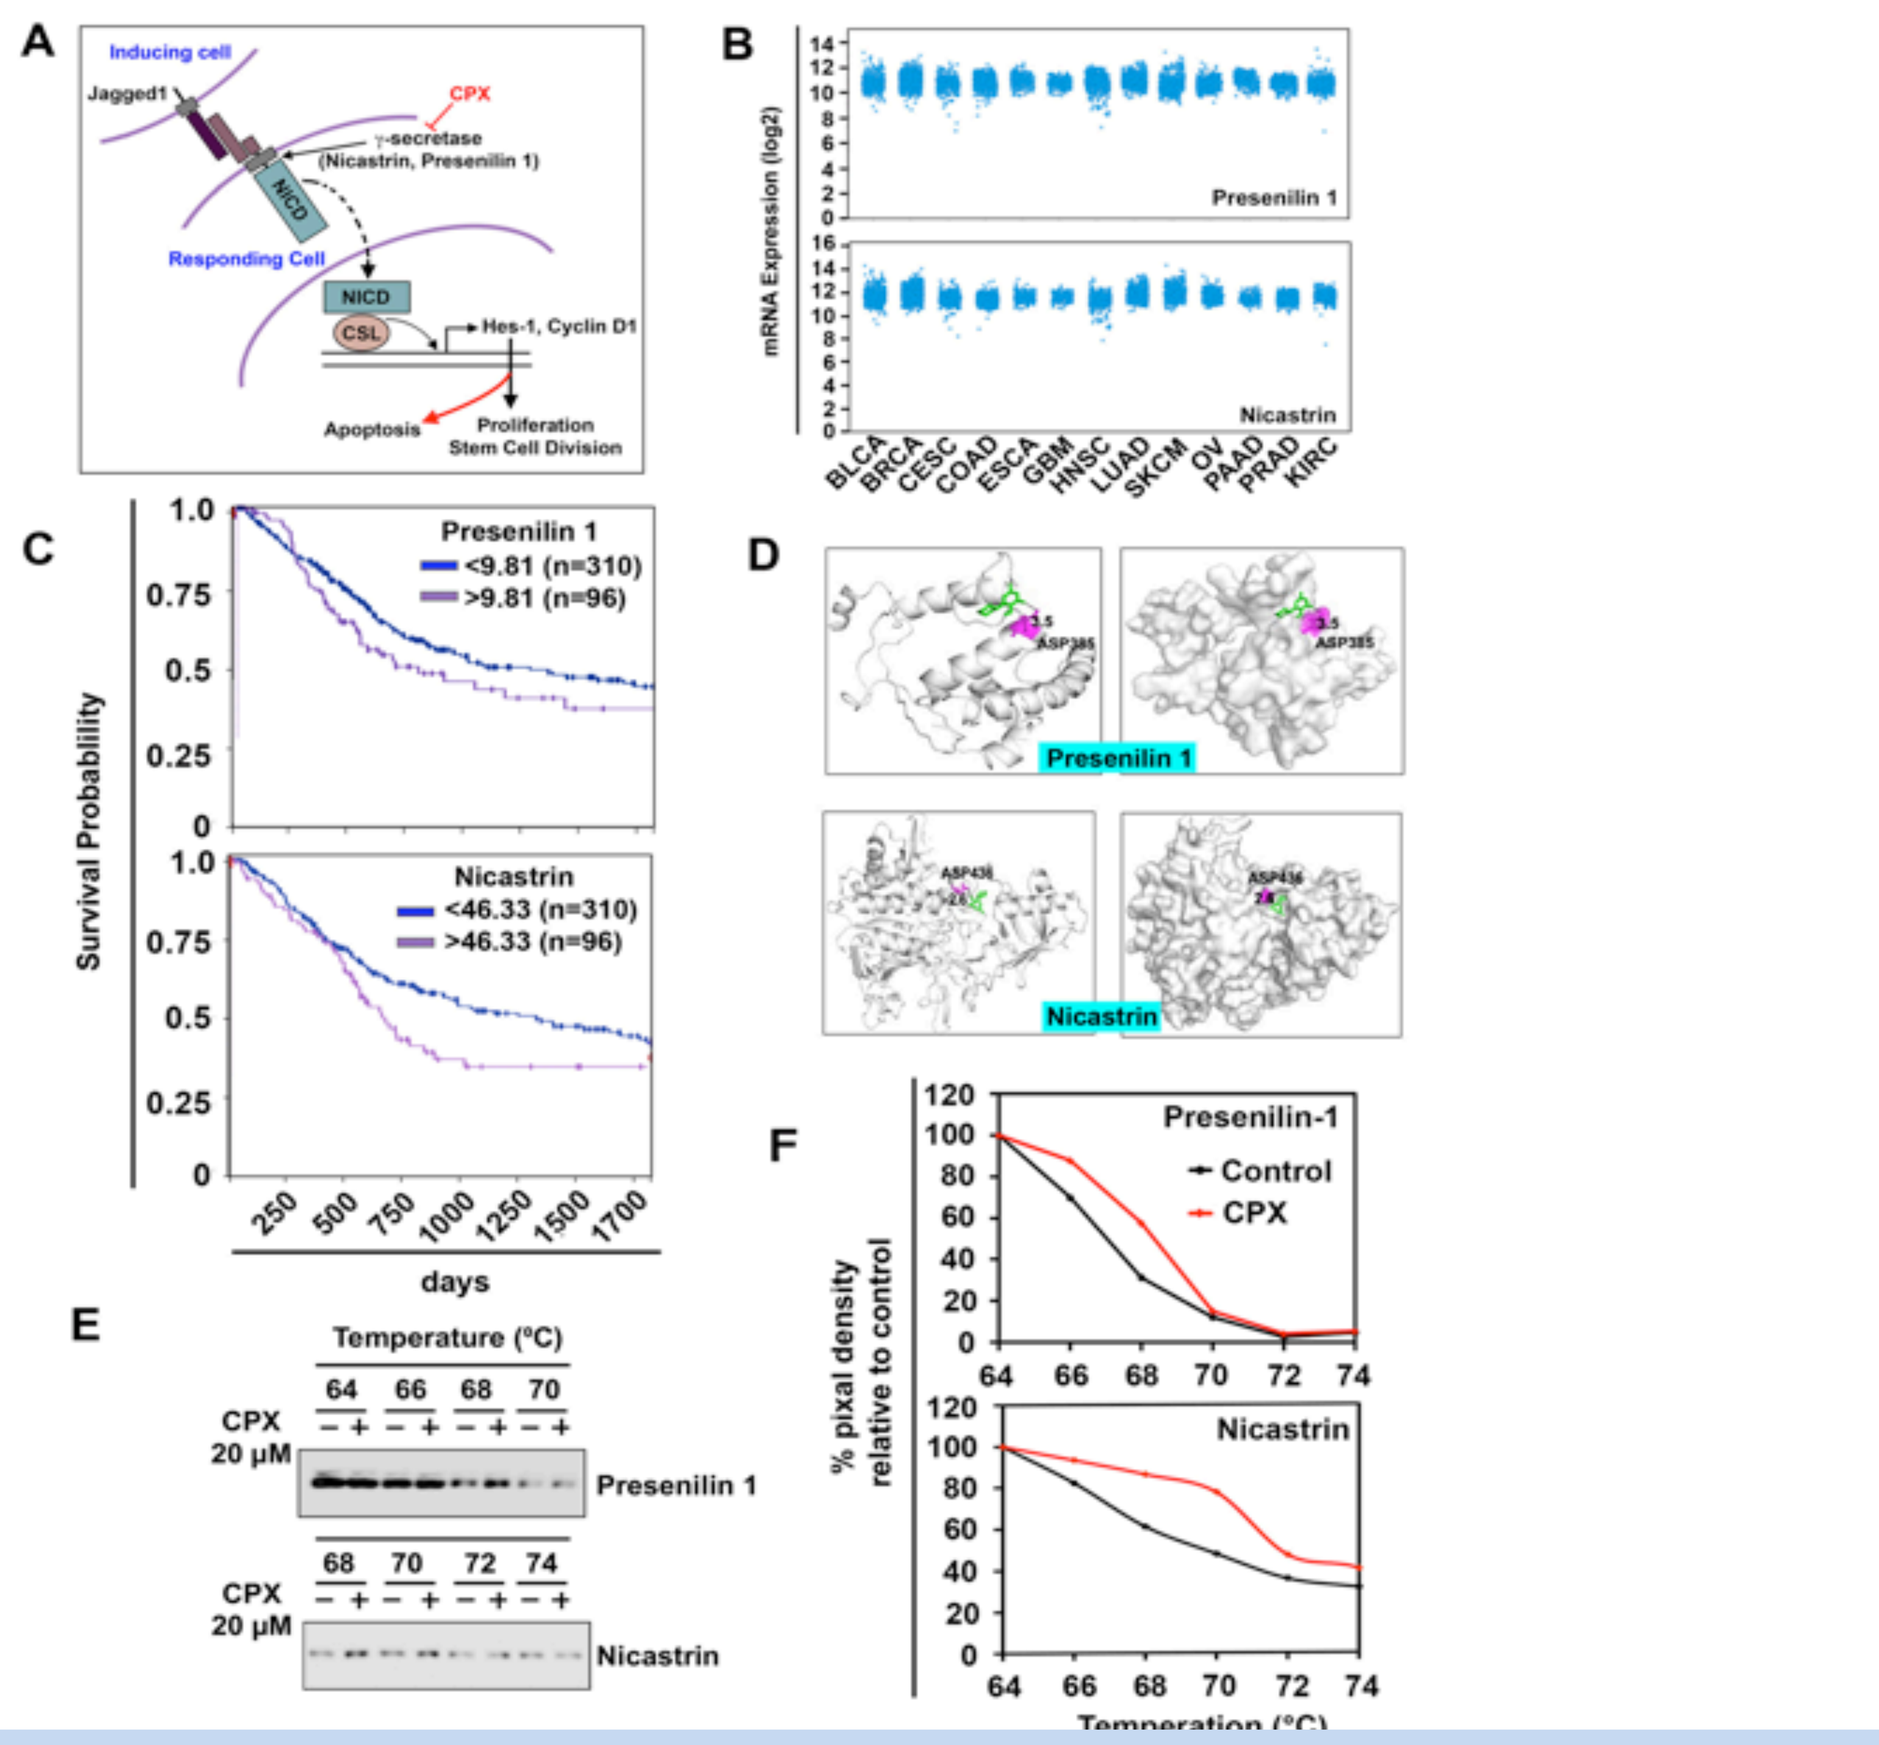
A
Inducing cell
Jagged1
CPX
γ-secretase (Nicastrin, Presenilin 1)
Responding Cell
NICD
CSL
Hes-1, Cyclin D1
Apoptosis
Proliferation Stem Cell Division
B
mRNA Expression (log2)
Presenilin 1
Nicastrin
BLCA BRCA CESC COAD ESCA GBM HNSC LUAD SKCM OV PAAD PRAD KIRC
C
Survival Probability
Presenilin 1
<9.81 (n=310)
>9.81 (n=96)
Nicastrin
<46.33 (n=310)
>46.33 (n=96)
days
D
E
Temperature (°C)
CPX 20 μM
F
% pixal density relative to control
Presenilin-1
Control
CPX

Figure 6. A. Data suggests CPX downregulates Notch by inhibiting γ-Secretase proteins Presenilin 1 and Nicastrin. **B.** By profiling the TCGA, Presenilin 1 and Nicastrin are higher in 13 cancer types. **C.** Kaplan-Meier survival analysis of the GEPIA database showed that Presenilin 1 and Nicastrin were upregulated in bladder cancer tissue. **D.** Molecular modeling showed CPX binds within the protein cavities of Presenilin 1 and Nicastrin. **E.** The cellular thermal shift assay (CETSA) confirmed CPX molecular docking results in T24 cells. F. Thermal denaturation of Presenilin 1 and Nicastrin in vehicle- and CPX-treated T24 cells demonstrated CPX protects the proteins from denaturation. Taken together, molecular docking and CETSA results demonstrate CPX binds with γ-secretase complex proteins.

[1]*Department of Cancer Biology, University of Kansas Medical Center, Kansas City, KS;* [2]*Department of Pharmacology, Toxicology and Therapeutics, University of Kansas Medical Center, Kansas City, KS;* [3]*Institute for Advancing Medical Innovation, University of Kansas Medical Center, Kansas City, KS;* [4]*Division of Medical Oncology, Department of Medicine, University of Kansas Medical Center, Kansas City, KS;* [5]*Department of Urology, University of Kansas Medical Center, Kansas City, KS;* [6]*Biopharmaceutics Innovation and Optimization Center, University of Kansas, Lawrence, KS;* [7]*Schoool of Pharmacy, Istanbul Kemerburgaz University, Istanbul, Turkey;* [8]*CicloMed LLC, Kansas City, MO;* [9]*Alba BioPharm Advisors Inc, Raleigh, NC;* [10]*The Gnomon Group LLC, Durham, NC;* [11]*Department of Pathology, University of Kansas Medical Center, Kansas City, KS*